Pharmacokinetic and Pharmacodynamic (PK/PD) Relationship of Intravenous Ganaxolone in Refractory Status Epilepticus

R. Eugene Ramsay, MD[4], Aatif M. Husain, MD[2], Christa B. Swisher, MD[2], Steven Smith[3], Heather Van Heusen[3], Julia Tsai, PhD[3], Lorianne Masuoka, MD[3], Maciej Gasior, MD, PhD[3], Henrikas Vaitkevicius, MD[1]

[1]Neurosciences Institute, Ochsner Health System, New Orleans, LA; [2]Dept. of Neurology, Duke University, Durham, NC; [3]Marinus Pharmaceuticals, Radnor, PA; [4]Dept. of Neurology, Brigham and Women's Hospital, Boston, MA

RSE OVERVIEW

Status Epilepticus (SE) is the most life-threatening occurrence within the spectrum of epileptic disorders

- Continuous seizures lasting >5 min for convulsive seizures or >30 minutes for non-convulsive seizures
- Heterogenous patient population with various/unknown etiologies
- Prolonged seizure activity can result in permanent neuronal damage and contribute to the high morbidity and mortality rates associated with SE
- SE becomes more difficult to control as its duration increases and is associated with increased mortality

Established status epilepticus (ESE) is defined as failure of at least one benzodiazepine at adequate doses	RSE defined as failure of at least one 2nd line intravenous (IV) anti-epileptic drug (AED)	Super Refractory Status Epilepticus (SRSE) is SE that continues or recurs 24hrs or more after the onset of IV anesthetics or failure to wean off IV anesthetics

GOALS OF A NEW THERAPY FOR THE TREATMENT OF RSE

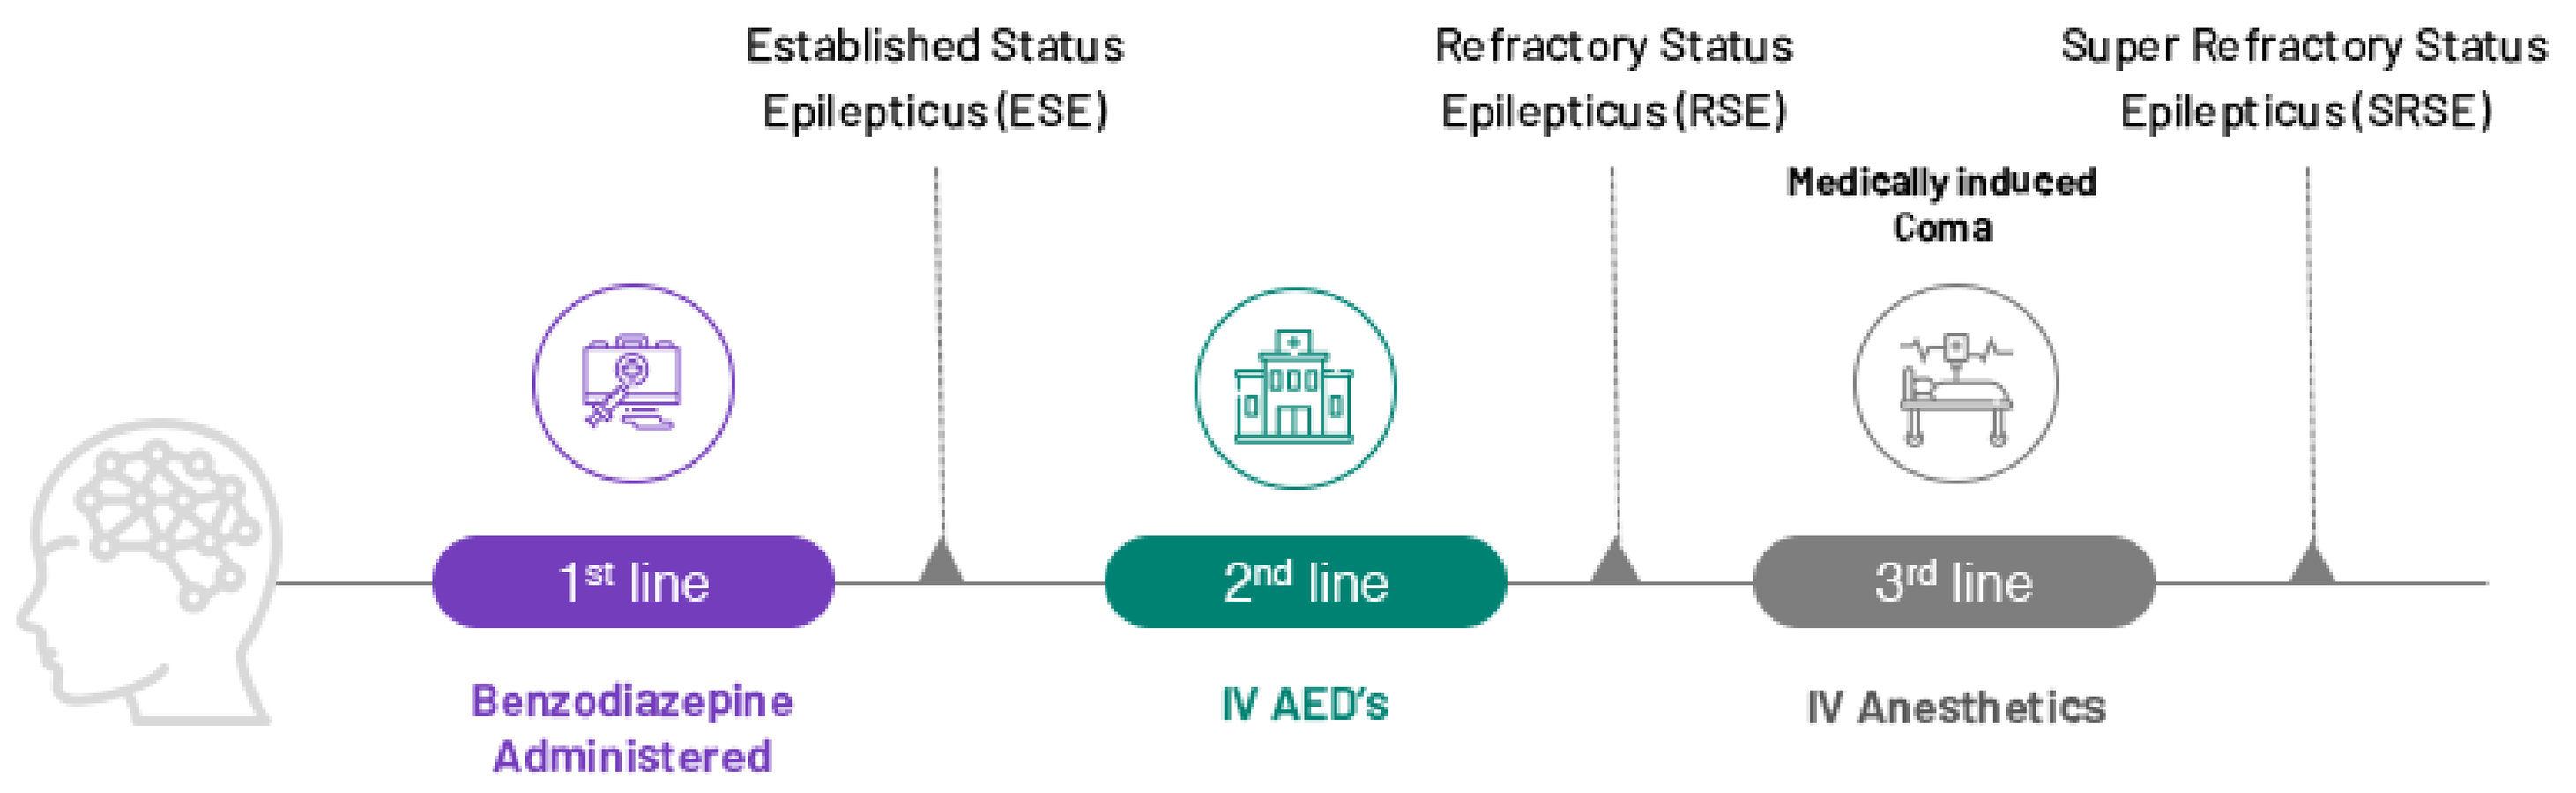
Established Status Epilepticus (ESE)
Refractory Status Epilepticus (RSE)
Super Refractory Status Epilepticus (SRSE)
Medically induced Coma
1st line
2nd line
3rd line
Benzodiazepine Administered
IV AED's
IV Anesthetics

Goals of a new treatment for RSE

Prevent patient progression towards escalation of treatment (IV anesthetics)

Rapid cessation of SE

Maintenance of seizure control over study period

GANAXOLONE (GNX) MECHANISM OF ACTION

- GNX is a synthetic analog of endogenous allopregnanolone and a potent positive allosteric modulator of $GABA_A$-receptors.

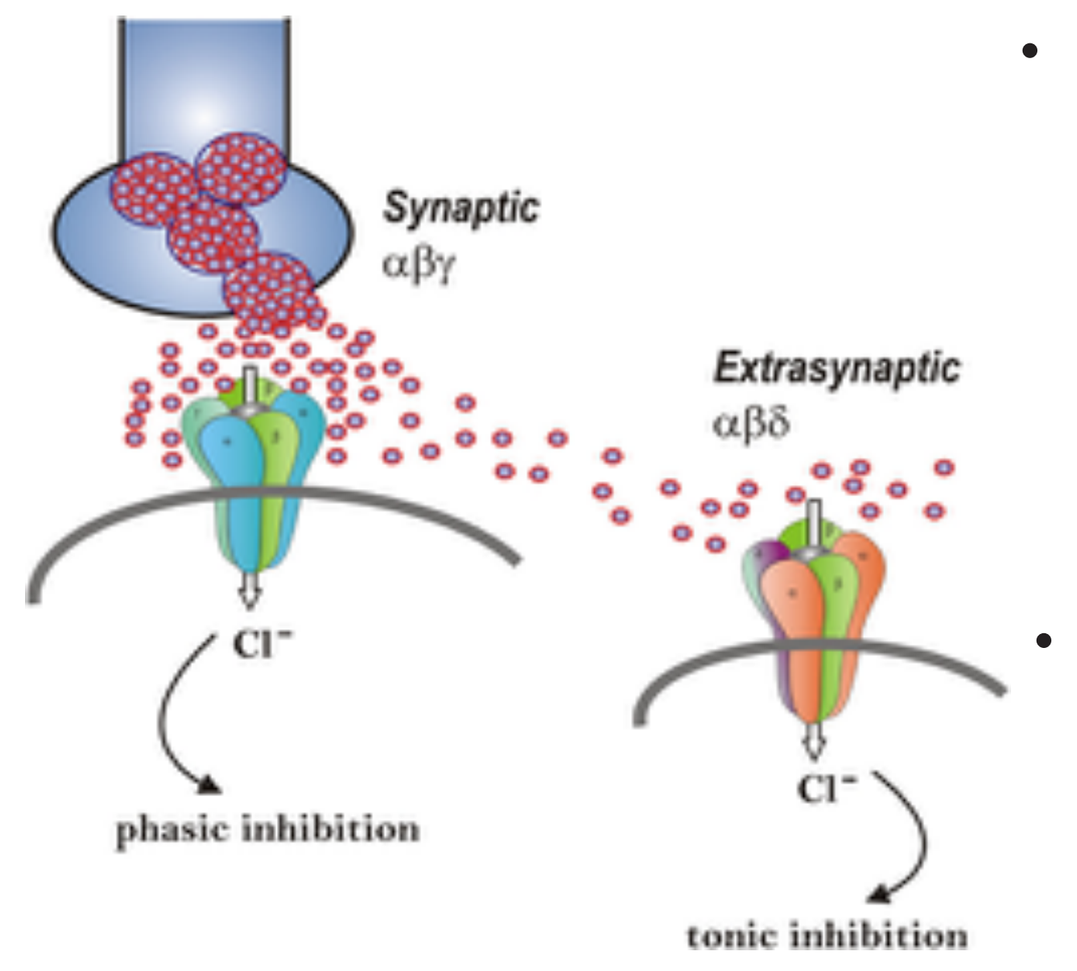
Synaptic
αβγ
Extrasynaptic
αβδ
Cl⁻
phasic inhibition
tonic inhibition

- GNX targets unique binding sites on $GABA_A$-receptors that are not susceptible to tolerance build up (e.g., benzodiazepines)

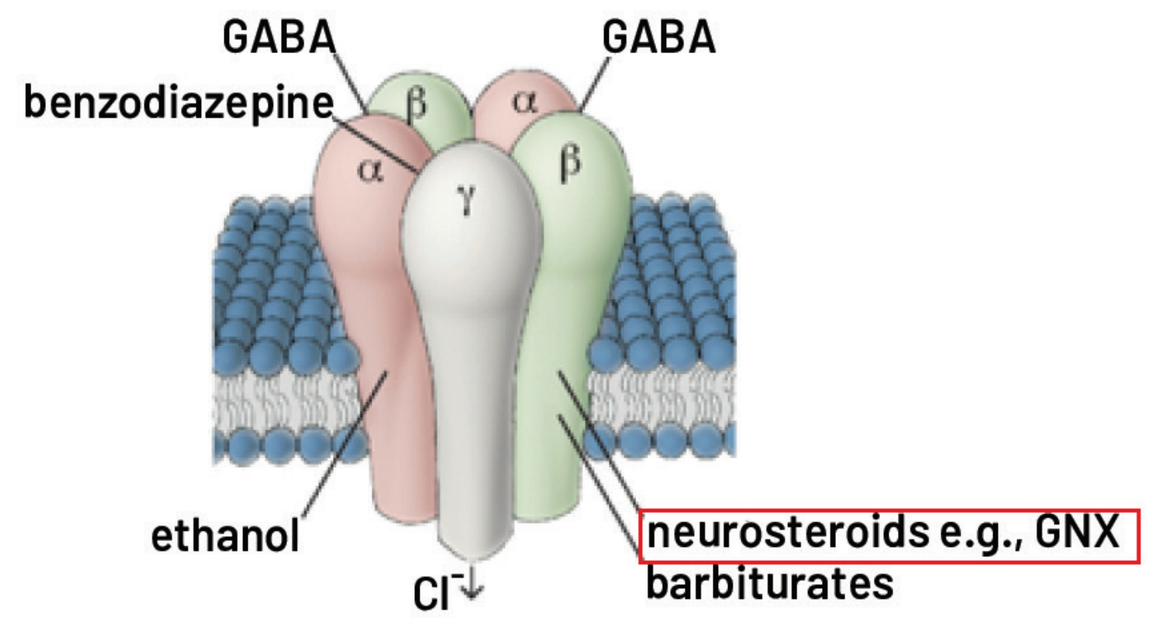
GABA
GABA
benzodiazepine
ethanol
Cl⁻
neurosteroids e.g., GNX
barbiturates

- GNX acts on both synaptic and extrasynaptic $GABA_A$-receptors to maximize inhibitory signaling as well as maintain activity when synaptic receptors are down-regulated

PHASE 2 TRIAL OF IV GANAXOLONE IN RSE

Study Design

Objectives: Evaluate safety, tolerability, efficacy, and pharmacokinetics of IV Ganaxolone in RSE patients

RSE Patients: Diagnosis of convulsive or non-convulsive SE; Failed at least one 2nd line IV AED but not progressed to 3rd line IV anesthetics

Screening → Treatment Period → Post-treatment Follow-up

Treatment Period		
Loading Dose	Maintenance	Taper
Bolus plus continuous infusion	2-4 day infusion	18 hour taper

Cohort	Dose of GNX/day	N
Low	500mg/day	5
Medium	650mg/day	4
Target	713mg/day	8

Post-treatment Follow-up	
24 hr	Weeks 2, 3, 4

Endpoints
- ***Primary:*** number of patients who do not require escalation of treatment with IV anesthetic within the first 24 hours after ganaxolone initiation
- ***Secondary:*** additional efficacy, safety and tolerability

Patient Baseline Characteristics

17 patients were enrolled (8 males, 9 females) with an average age of 57 years old (range: 23-88)

Patient	Dosing Cohort	Etiology	History of Epilepsy	Type of SE	Failed Antiseizure Medications Prior to GNX*
1	Low	Vascular	No	NCSE	LAC, LEV
2	Low	Unknown	Yes	NCSE	fPHT, LEV
3	Low	Vascular	No	NCSE	LOR, LAC, LEV
4	Low	Vascular	No	NCSE	LOR, LAC, LEV
5	Low	Tumor	No	CSE	LOR, LAC, LEV
6	Medium	Vascular	No	NCSE	LOR, LAC, LEV
7	Medium	Drug Overdose / Withdrawl	Yes	CSE	LOR, LEV
8	Medium	Unknown	Yes	CSE →NCSE	LOR, LAC, LEV
9	Medium	Tumor	Yes	NCSE	LAC, LEV, PHT
10	Target	Vascular	Yes	CSE	LOR, LAC, VPA
11	Target	Drug Overdose / Withdrawl	No	CSE	LOR, LAC, LEV
12	Target	Tumor	Yes	NCSE	LOR, LEV, VPA
13	Target	Autoimmune	No	NCSE	LOR, LEV
14	Target	Vascular	No	NCSE	LOR, LAC, LEV, PHT
15	Target	Vascular	Yes	CSE	LOR, LEV
16	Target	Tumor	No	NCSE	LOR, LAC, LEV
17	Target	Autoimmune	No	NCSE	LOR, fPHT, LAC, LEV, VPA

NCSE: Non-convulsive status epilepticus, CSE: Convulsive status epilepticus, LAC: Lacosamide, LEV: Levetiracetam, LOR: Lorazepam, PHT: Phenytoin, fPHT: Fosphenytoin, VPA: Valproic Acid
*All prior AED's were administered at therapeutically relevant doses and within recommended dosing guidelines

Efficacy of IV Ganaxolone

Cohort	No escalation to IV anesthetics within 24 hours from infusion initiation **(Primary Endpoint)**	Status-free through 24 hours from infusion initiation	No escalation to additional IV AEDs or IV anesthetics for status relapse at any time through 24hr after GNX discontinuation	No SE Relapse at anytime during the 4-wk follow up period	
Target (713 mg/day; n=8)	100% (8 of 8)	88% (7 of 8)	100% (8 of 8)	100% (6 of 6); *1ET, 1 died*	One patient had status relapse @ Day 1, which resolved during the ganaxolone infusion without treatment escalation
Medium (650 mg/day; n=4)	100% (4 of 4)	100% (4 of 4)	75% (3 of 4)	67% (2 of 3); *1 ET*	One patient escalated to additional IV AED @ Day 1 for seizure relapse. One patient experienced status relapse @ Day 2 (during taper).
Low (500 mg/day; n=5)	100% (5 of 5)	100% (5 of 5)	60% (3 of 5)	50% (1 of 2); *1 died*	Two patients escalated to 3rd line therapy for seizure relapse @ Day3

ET: Early Termination

Safety of IV Ganaxolone

10 SAEs in 6 patients (also included in AEs)	50 AEs in 16 patients
2 related in 2 patients • 2 severe sedation	**13 Related in 7 subjects** • 6 mild (2 hypotension, 2 somnolence, 1 urinary retention, 1 hypercarbia) • 5 moderate (4 somnolence; 1 hypercarbia) • 2 severe (2 sedation)

Intubation

9 patients were not intubated upon enrollment. Of these, 6 remained intubation-free during the entire ganaxolone treatment period

PK/PD Relationship and Rationale for Target Dose

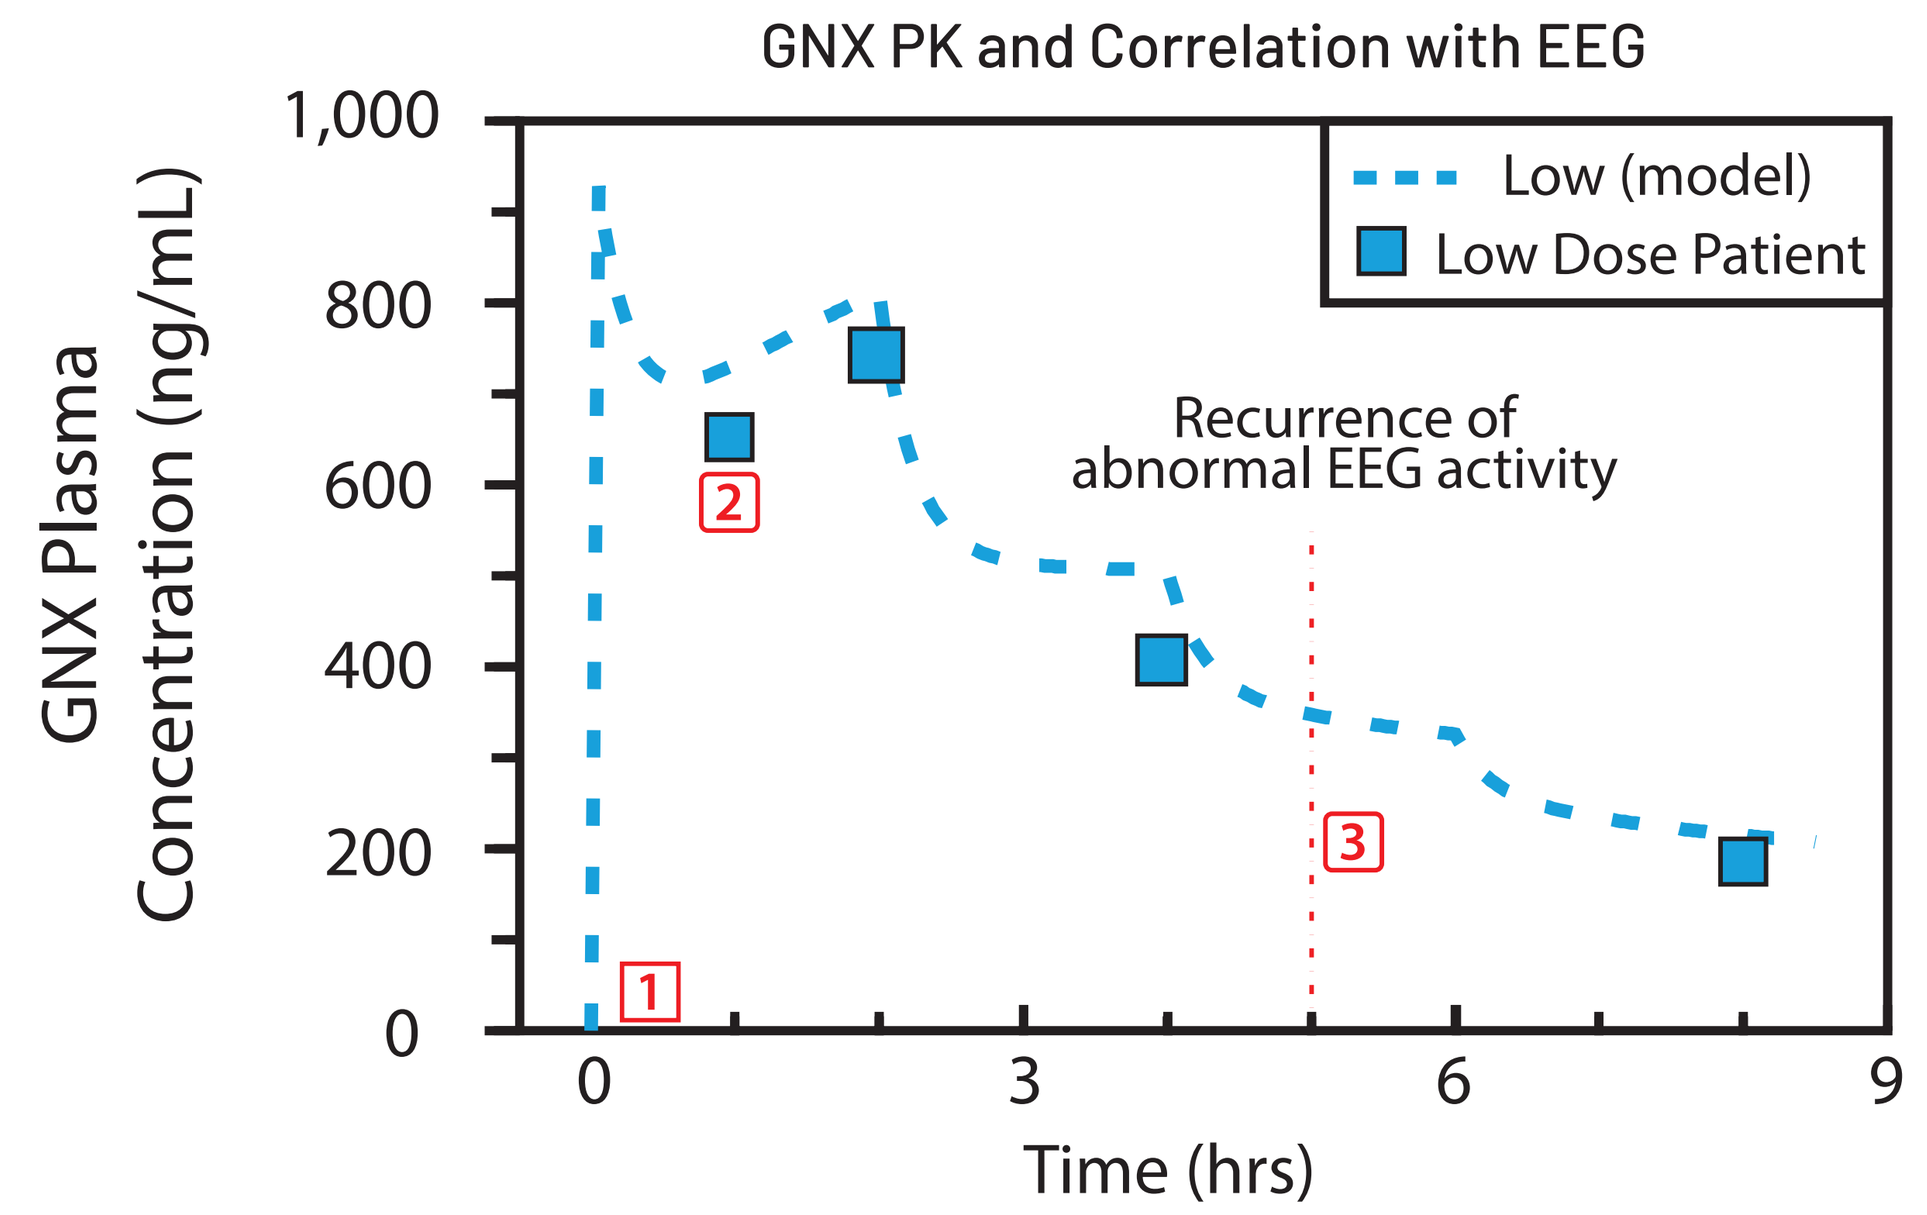
GNX PK and Correlation with EEG
GNX Plasma Concentration (ng/mL)
1,000
800
600
400
200
0
0
3
6
9
Time (hrs)
Low (model)
Low Dose Patient
Recurrence of abnormal EEG activity
1
2
3

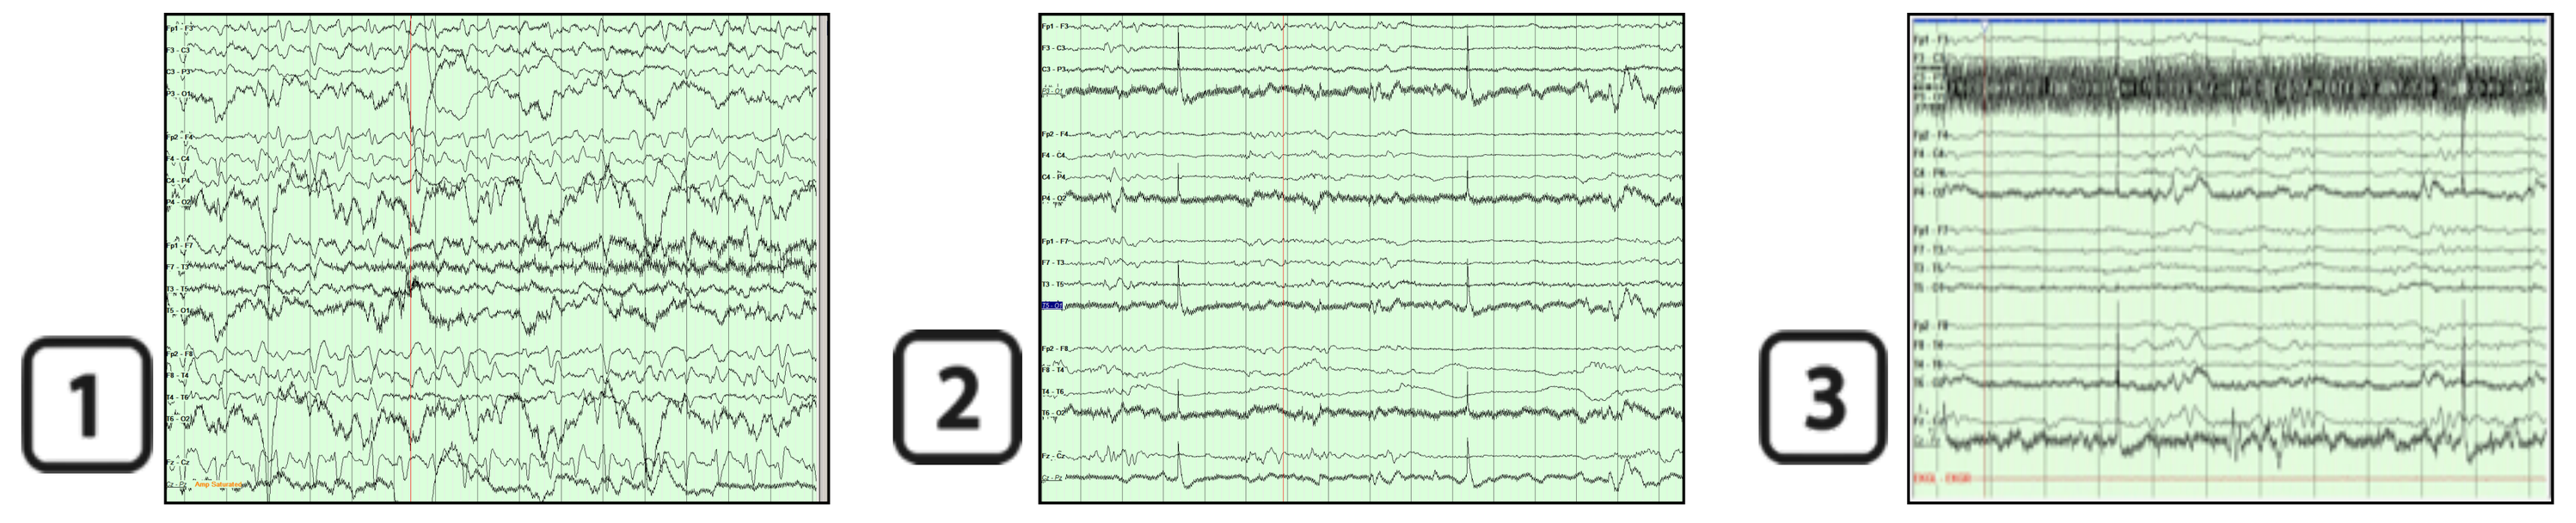
1
2
3

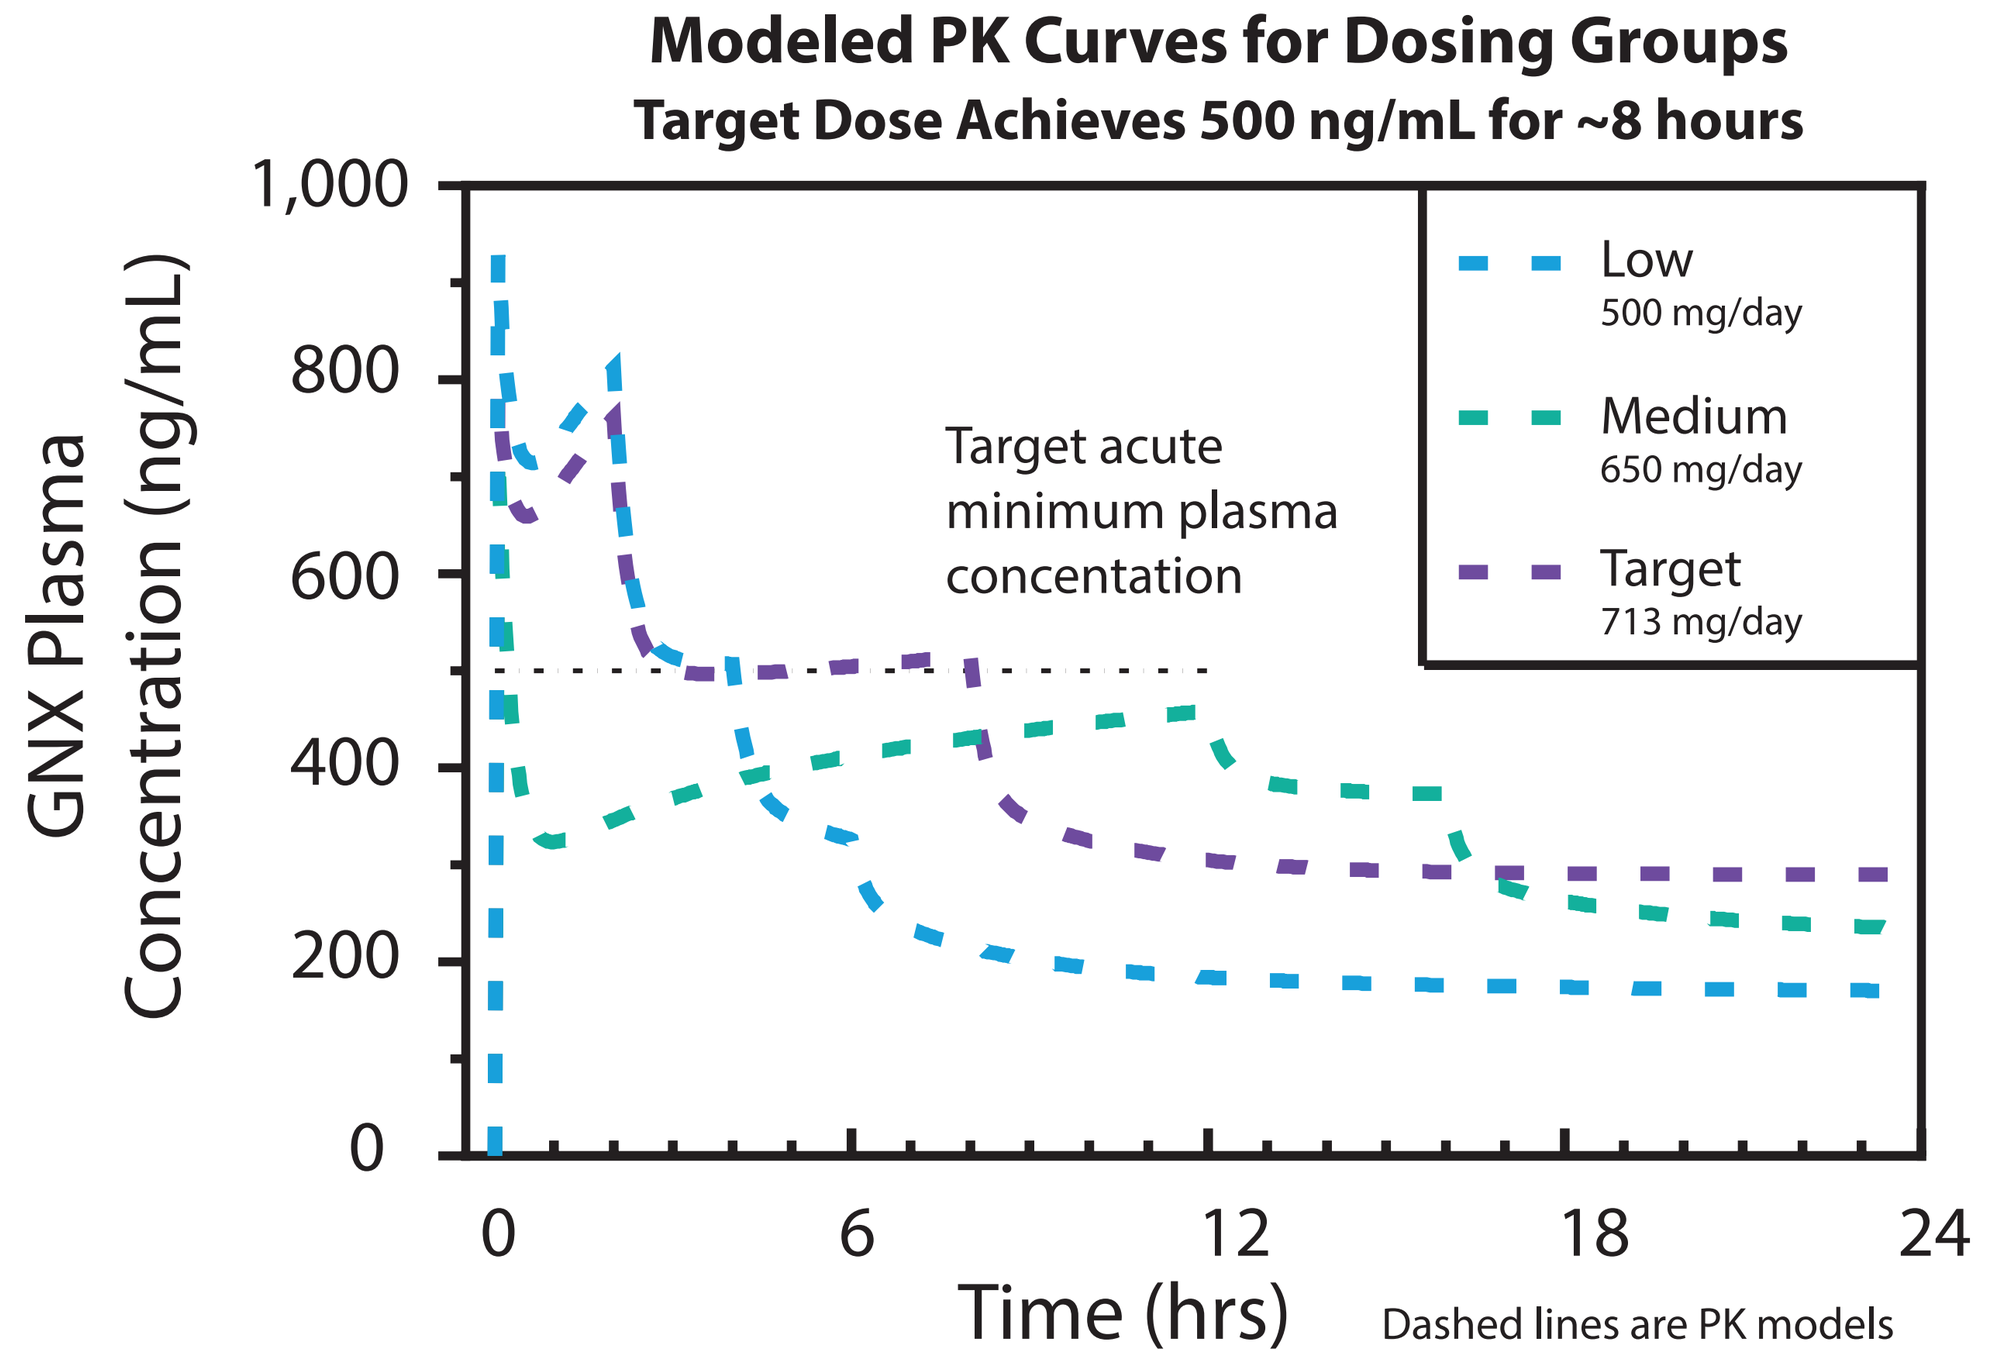
Modeled PK Curves for Dosing Groups
Target Dose Achieves 500 ng/mL for ~8 hours
GNX Plasma Concentration (ng/mL)
1,000
800
600
400
200
0
0
6
12
18
24
Time (hrs)
Target acute minimum plasma concentation
Low
500 mg/day
Medium
650 mg/day
Target
713 mg/day
Dashed lines are PK models

Conclusions

- No patients progressed to IV anesthetics during first 24 hours
 Median time to SE cessation = 5 minutes (3.4-14.4 minutes 25th-75th percentile) (n=15 evaluable)
 Durable response throughout study period in target dose cohort
- Patients failed mean of 2.1 second line IV AEDs
 Highly heterogeneous underlying cause of status
- Target patient population and dose identified for Ph. 3 study
- Ganaxolone shows an acceptable safety profile in patients with RSE