#209

IV Ganaxolone in Pediatric Super Refractory Status Epilepticus: A Single Patient Case Study

Rajdeep Singh, MD[1]; Stephanie Robinett, MD[2]; Alejandra Stewart, MD[2]; Katherine Van Poppel, MD[2]; Joseph Hulihan, MD[3]; Maciej Gasior, MD, PhD[3]; Rani Singh, MD[2]

[1]Atrium Health Neurosciences Institute, Charlotte, NC, USA; [2]Department of Pediatrics, Atrium Health Levine Children's Hospital, Charlotte, NC, USA;
[3]Marinus Pharmaceuticals, Inc., Radnor, PA, USA

Introduction

- Super refractory status epilepticus (SRSE) is defined as status epilepticus (SE) that continues for 24 hours or more despite anesthetic treatment or that recurs on an attempted wean of the anesthetic regimen[1]
 - SRSE is a life-threatening neurological emergency associated with significant morbidity and mortality[2,3]
- Although epidemiological studies on SRSE are based mainly on adult populations, a retrospective study showed ≈7% SRSE occurrences in children with convulsive SE episodes[4]
- High-quality comparative clinical studies of various SRSE treatment strategies have not been conducted
 - Treatment decisions mainly rely on case series or expert opinion[1,5]
- Ongoing seizures are known to cause internalization of synaptic γ-amino butyric acid type A ($GABA_A$) receptors playing a key role in the development of pharmacoresistance.[6,7] The function and number of extrasynaptic $GABA_A$ receptors in prolonged SE have been found to be preserved[7,8] and identified as a potential mechanism for therapeutic intervention

Potential role of neuroactive steroids

- Neuroactive steroids that act as positive $GABA_A$ receptor modulators exhibit broad-spectrum anticonvulsant effects and may play a role in regulating epileptogenesis[9]
- PCDH19-related epilepsy has been associated with low levels of allopregnanolone and potential GABAergic dysfunction[10]
- Ganaxolone (GNX) is a synthetic analog of allopregnanolone and a potent positive allosteric modulator of $GABA_A$ receptors
- GNX acts on both synaptic and extrasynaptic $GABA_A$ receptors to maximize inhibitory signaling as well as maintain activity when synaptic receptors are downregulated

GNX clinical data in refractory status epilepticus (RSE)

- An open-label, dose-finding phase 2 study evaluating intravenous (IV) GNX in 17 patients with RSE demonstrated:
 - Median time to SE cessation of 5 minutes and no patients progressing to IV anesthetics within 24 hours of treatment initiation
 - GNX had an acceptable safety and tolerability profile for the RSE patient population in all dose groups
- Currently, IV GNX is being studied in a phase 3, randomized, double-blind study in addition to standard-of-care antiseizure medications (ie, 2nd-line IV antiepileptic drugs [AEDs]) and before IV anesthetic during the treatment of refractory SE (NCT04391569)
- There are no published studies with GNX in SRSE

Case Presentation

- We discuss a 17-year-old female who had been hospitalized for 8 months with recurrent RSE and who relapsed while in our inpatient rehabilitation unit. She was treated with IV GNX (Marinus Pharmaceuticals, Inc.) under an emergency investigational new drug (E-IND) application to the FDA

Clinical history and course

- History of sporadic, primarily febrile and rare afebrile convulsive seizures in early childhood; seizure-free for 10 years off medication until she began to have focal seizures with generalization. She had recurrent seizures over the first 3 weeks and was admitted to an outside hospital for SE requiring intubation
- Over the course of 7 months at the initial tertiary hospital, she had 6 episodes of SE and required intubation 4 times with medically induced coma for seizure suppression. Prior AEDs and other key treatments are summarized in **Table 1**. She was transferred to our inpatient pediatric rehabilitation unit after being seizure-free for 1 month on 5 antiseizure medications (cannabidiol, perampanel, phenobarbital, lacosamide, lorazepam), pyridoxine, ketogenic diet, anakinra, and menstrual suppression
- A thorough infectious, metabolic, genetic, vascular, and autoimmune evaluation at both institutions was nondiagnostic (**Figure 1**)
- While in the inpatient rehabilitation unit, the patient developed a fever of 41.7°C, and a respiratory viral panel was positive for parainfluenza. SE returned, requiring transfer to the intensive care unit for midazolam and pentobarbital infusions
- Video-electroencephalogram (vEEG) monitoring throughout her course demonstrated multifocal seizure onset. She was seizure-free for 2 days after drips were weaned, until seizures returned, prompting placement of a vagus nerve stimulator and a prolonged 2-week course of IV pentobarbital titrated to EEG burst suppression. A repeat magnetic resonance imaging demonstrated progression of diffuse global parenchymal atrophy (**Figure 2**). As pentobarbital was weaned off, convulsive seizures returned (**Table 2**)
- She was treated under the E-IND approval for a trial of IV-to-enteric GNX. GNX was administered using an IV bolus, followed by infusion over 4 days (with GNX boluses as needed for breakthrough seizures) (**Figure 3**)
 - On day 1, pentobarbital was discontinued; by day 3, clinical and electrographic seizures stopped
 - The patient tolerated the protocol well. As pentobarbital was discontinued and GNX treatment initiated, she became more alert and interactive
 - On day 5 she was transitioned from IV to enteric GNX and has remained seizure-free

Patient case update: post discharge

- One month after GNX initiation, the patient was transferred to inpatient rehabilitation service; she was discharged home 1 month later. She has been weaned off the ketogenic diet, anakinra, and felbamate with minimal breakthrough clinical seizures (only in setting of intercurrent illness)
- Three months later, the patient's response to GNX and seizure cessation has been sustained

Table 1. Prior Treatments at OSH

Prior AEDs and therapies	Notes
Clobazam	Switched to lorazepam when PLEX initiated
Oxcarbazepine, then switched to carbamazepine	Weaned because of drug-drug interactions
Fosphenytoin	Worsened seizures
Topiramate	Ineffective
CBD oil	Ineffective
Leucovorin	
Ketamine gtt	Unsuccessful in suppressing seizures
Propofol gtt	Elevated CK; no other signs of propofol toxicity
Pentobarbital gtt	Responded well consistently throughout her admission
Nexplanon® (etonogestrel implant) and Supprelin®LA (histrelin acetate)	Implanted because of concern for seizures starting 4-7 days prior to menstrual cycle starting and catamenial trigger
IV Solu-Medrol® and IVIG	Unclear if beneficial
PLEX	EEG reportedly improved after PLEX initiated
Rituximab	
Kineret® (anakinra)	Started on April 23, 2020, for intractable epilepsy with "significant improvement in seizure frequency, interictal, clinical, and subclinical seizures"

AED, antiepileptic drugs; CBD, cannabidiol; CK, creatine kinase; EEG, electroencephalogram; gtt, drops; IV, intravenous; IVIG, intravenous immune globulin; OSH, outside hospital; PLEX, plasma exchange.

Figure 1. Diagnostic Studies

Infectious	Autoimmune/inflammatory	Metabolic
B cell panel: negative HLH gene panel: negative HIV and hepatitis: negative COVID Ag testing ×3: negative	CSF 4/2/20: 1 cell, 57 protein, 71 glucose Autoimmune encephalopathy panel: negative Pelvic US: negative for teratoma Oligoclonal band panel: negative NMO: negative	Serum carbohydrate deficient transferrin: essential normal, not consistent with CDG Serum biotinidase level, 13.8 U/L, within normal limits Homocysteine: low 3.5 Urine purine and pyrimidine panel: normal Urine sulfocysteine: normal Urine mucopolysaccharides screen: with elevated heparan sulfate, 0.33 Serum carbohydrate deficient transferrin: not consistent with congenital disorders of glycosylation Acylcarnitine profile: normal Free and total L-carnitine: plasma level normal Urine organic acids: normal Plasma amino acids: essentially normal CSF metabolic studies: negative

Imaging	Genetic
Normal unenhanced and enhanced MRI of the brain (×5) Negative MRA of the head	Epilepsy panel[a] with 5 VUS: PCDH19, POLG, DOCK7, NRXN1, and PLCB1 c.39-5T>G DOCK7 gene: ✓ father; X mother c.2299A>G p.Met767Val NRXN1 gene: ✓ mother; X father c.199A>C p.Ser67Arg PLCB1 gene: ✓ father; X mother c.3131T>C p.Val1044Ala POLG gene: ✓ father; X mother All are responsible for autosomal recessive disorders, and a second change was not found c.545G>C pGly182Ala PCDH19 gene: ✓ mother; X father Mother asymptomatic; uncertain significance Whole exome sequencing with mitochondrial genome (TRIO): negative

Ag, antigen; CDG, congenital disorders of glycosylation; COVID, coronavirus disease; CSF, cerebrospinal fluid; HLH, hemophagocytic lymphohistiocytosis; MRA, magnetic resonance angiography; MRI, magnetic resonance imaging; NMO, neuromyelitis optica; US, ultrasound.
[a]GeneDx, Inc., Gaithersburg, MD.

Figure 2. MRI During Prolonged Status Epilepticus

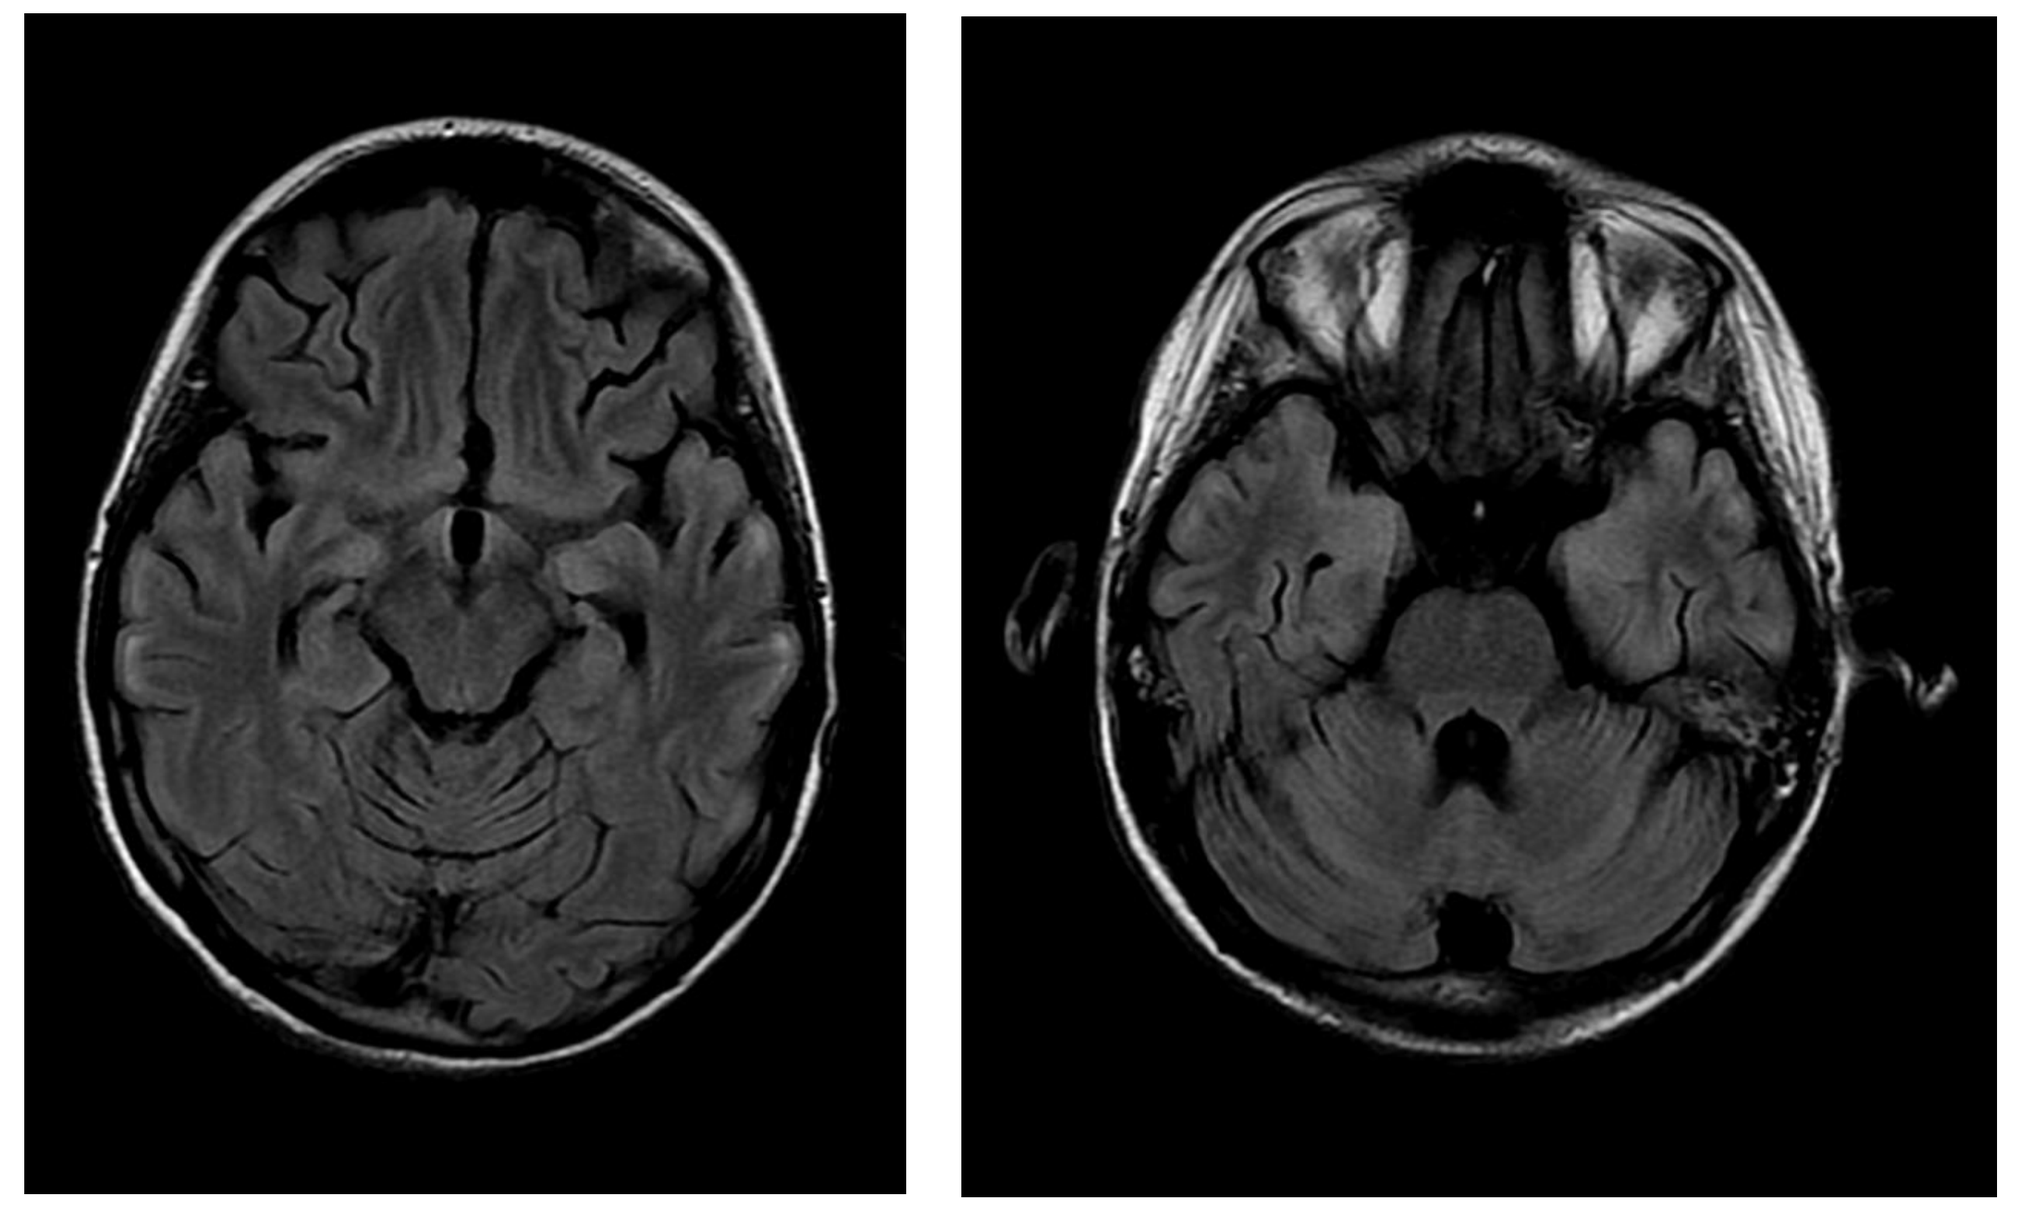

Marked progression of diffuse global parenchymal atrophy since 5 months prior, predominantly affecting the white matter. Increasing T2 FLAIR hyperintensity in the subcortical and juxta cortical white matter. Signal abnormality is most pronounced in the U fibers of the parietal and temporal lobes.
MRI, magnetic resonance imaging; T2 FLAIR, T2-weighted fluid-attenuated inversion recovery.

Figure 3. GNX Dosing

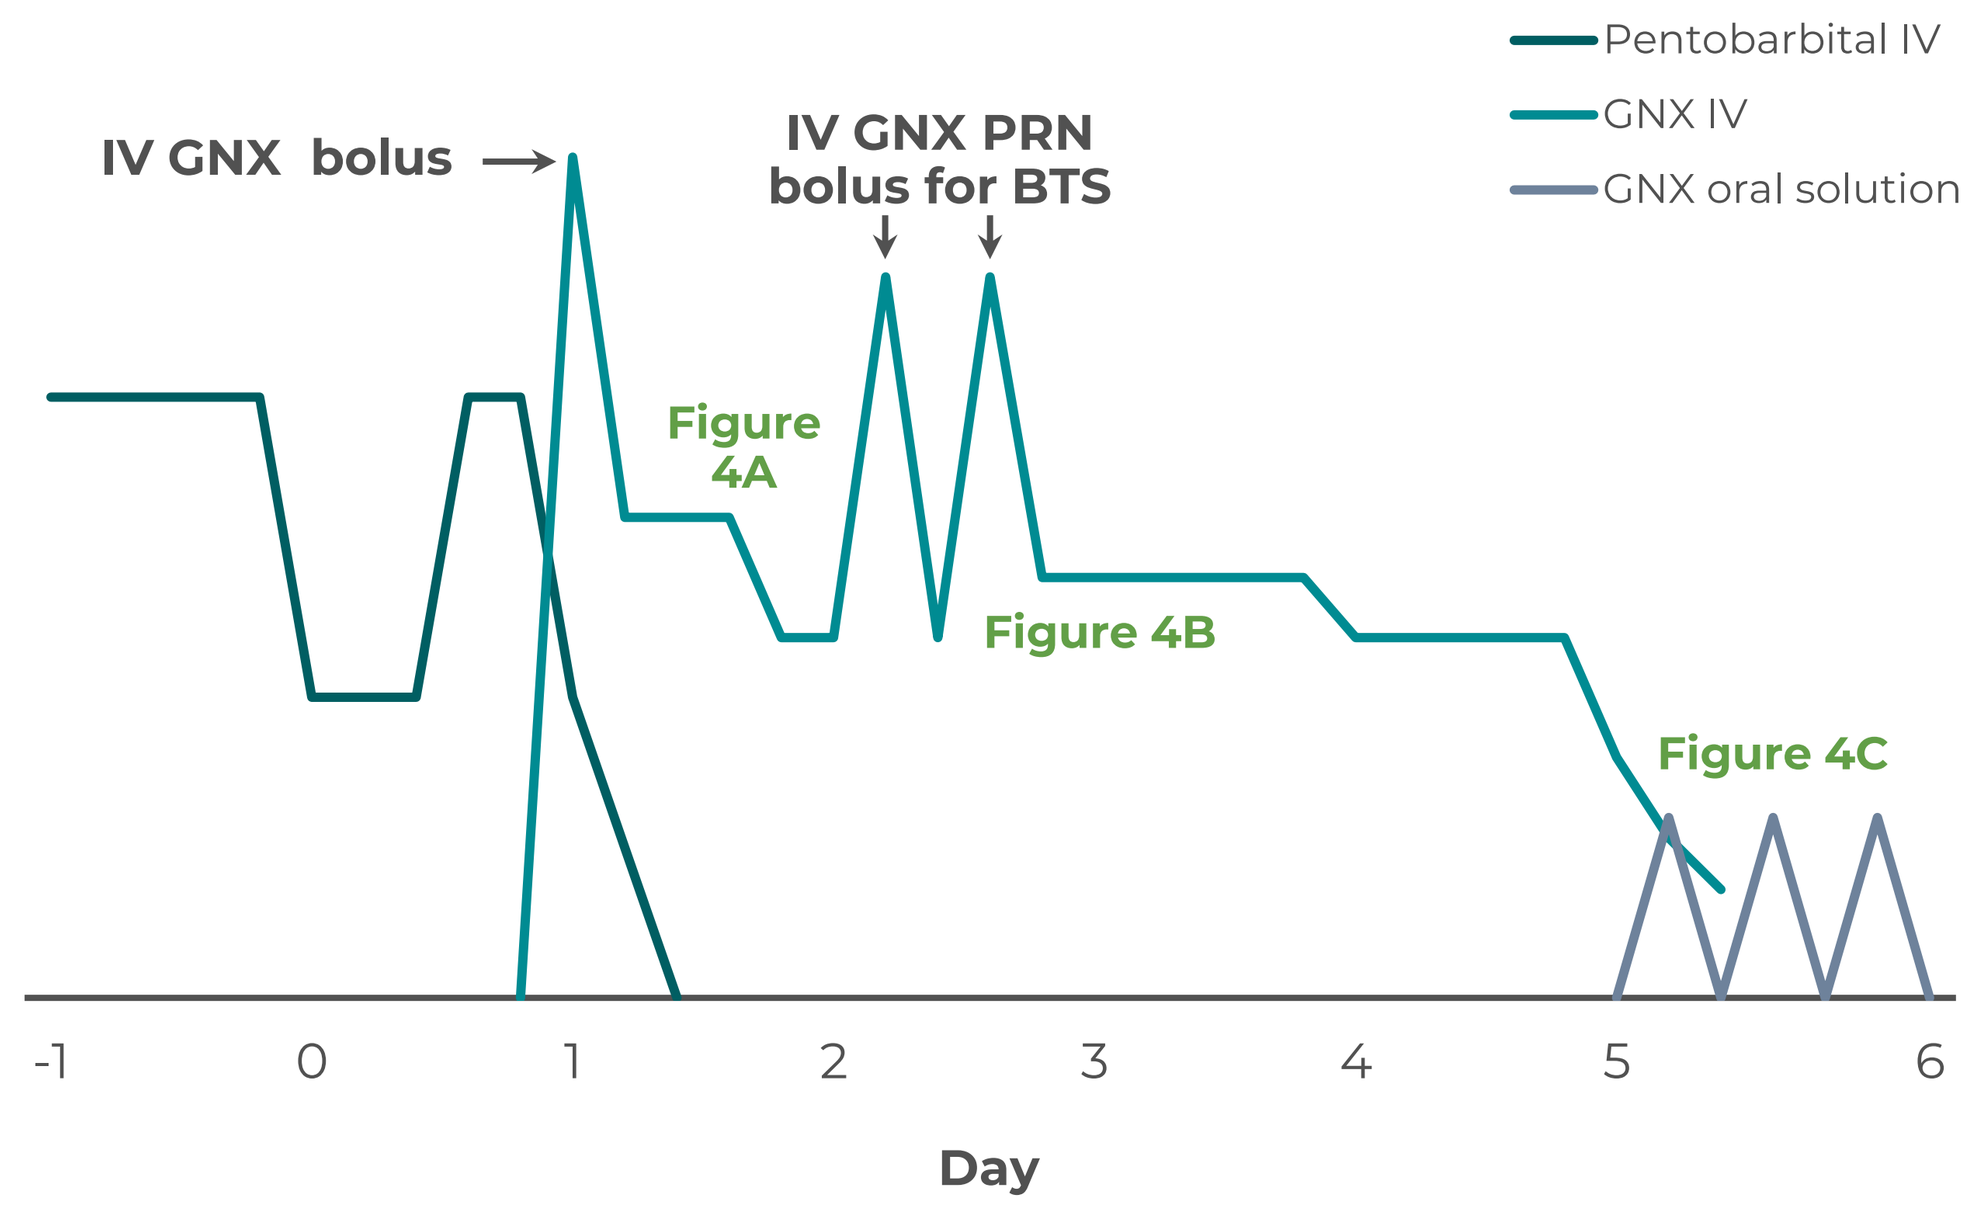
Pentobarbital IV
GNX IV
GNX oral solution
IV GNX bolus
IV GNX PRN bolus for BTS
Figure 4A
Figure 4B
Figure 4C
-1
0
1
2
3
4
5
6
Day

Pentobarbital mg/kg/hr converted to mg/hr. Ganaxolone CI dosed in mg/hr. Reference points for Figures 4A, 4B, and 4C are shown in green. BTS, breakthrough seizures; CI, continuous infusion; GNX, ganaxolone; IV, intravenous; PRN, as needed.

Table 2. Seizure Timeline Prior to and During GNX Administration

Day of therapy in relation to IV GNX initiation	Day −1	Day 0	Day 1	Day 2	Day 3	Day 4	Day 5
EEG seizures	9:00 AM, 1:00 PM, 7:00 PM, 8:00 PM <30 s each, marked by evolution of rhythmic spike and wave activity in the R temporal region T4, T6, sometimes also in the R frontal region	1:00 AM, 2:00 AM, 3:00 AM Similar to EEG seizures on day −1	3:00 PM (×2), 7:00 PM, 9:00 PM 5-25 s each; evolution of high-amplitude spike, wave discharges over the R temporal region, T4 T6 to C4. No clinical correlate	5:00 AM, 8:00 AM, 9:00 AM (×2), 10:00 AM, 12:00 PM Sporadic, isolated, subtle, self-limited; stopped once the secondarily generalized seizures stopped Same as EEG seizures on day 1	8:00 AM ≈10 s; evolution of high-amplitude spike, wave discharges over the R temporal region, T4 T6	2:00 AM ≈30 s; low-amplitude, fast activity over the R posterior quadrant	
Clinical and EEG seizures with minimal clinical correlate		7:00 AM, 9:00 AM (×3); 10:00 AM, 12:00 PM <30 s each; EEG with evolution of high-amplitude spike, wave discharges over the R T4, T6 to C4; difficult to assess; sometimes with some oral automatisms					
Clinical and EEG seizures with generalization requiring treatment		12:00 PM, 2:00 PM GTC seizures; EEG, ≈3 min		1:00 PM, 7:00 PM Focal seizures that generalized	Similar to focal generalized seizures as on day 2		

Times are estimated to the closest full hour. C, central; EEG, electroencephalogram; GNX, ganaxolone; GTC, generalized tonic-clonic; IV, intravenous; PTB, pentobarbital; R, right; T, temporal.

Figure 4A. Longitudinal Bipolar Montage—Day 1

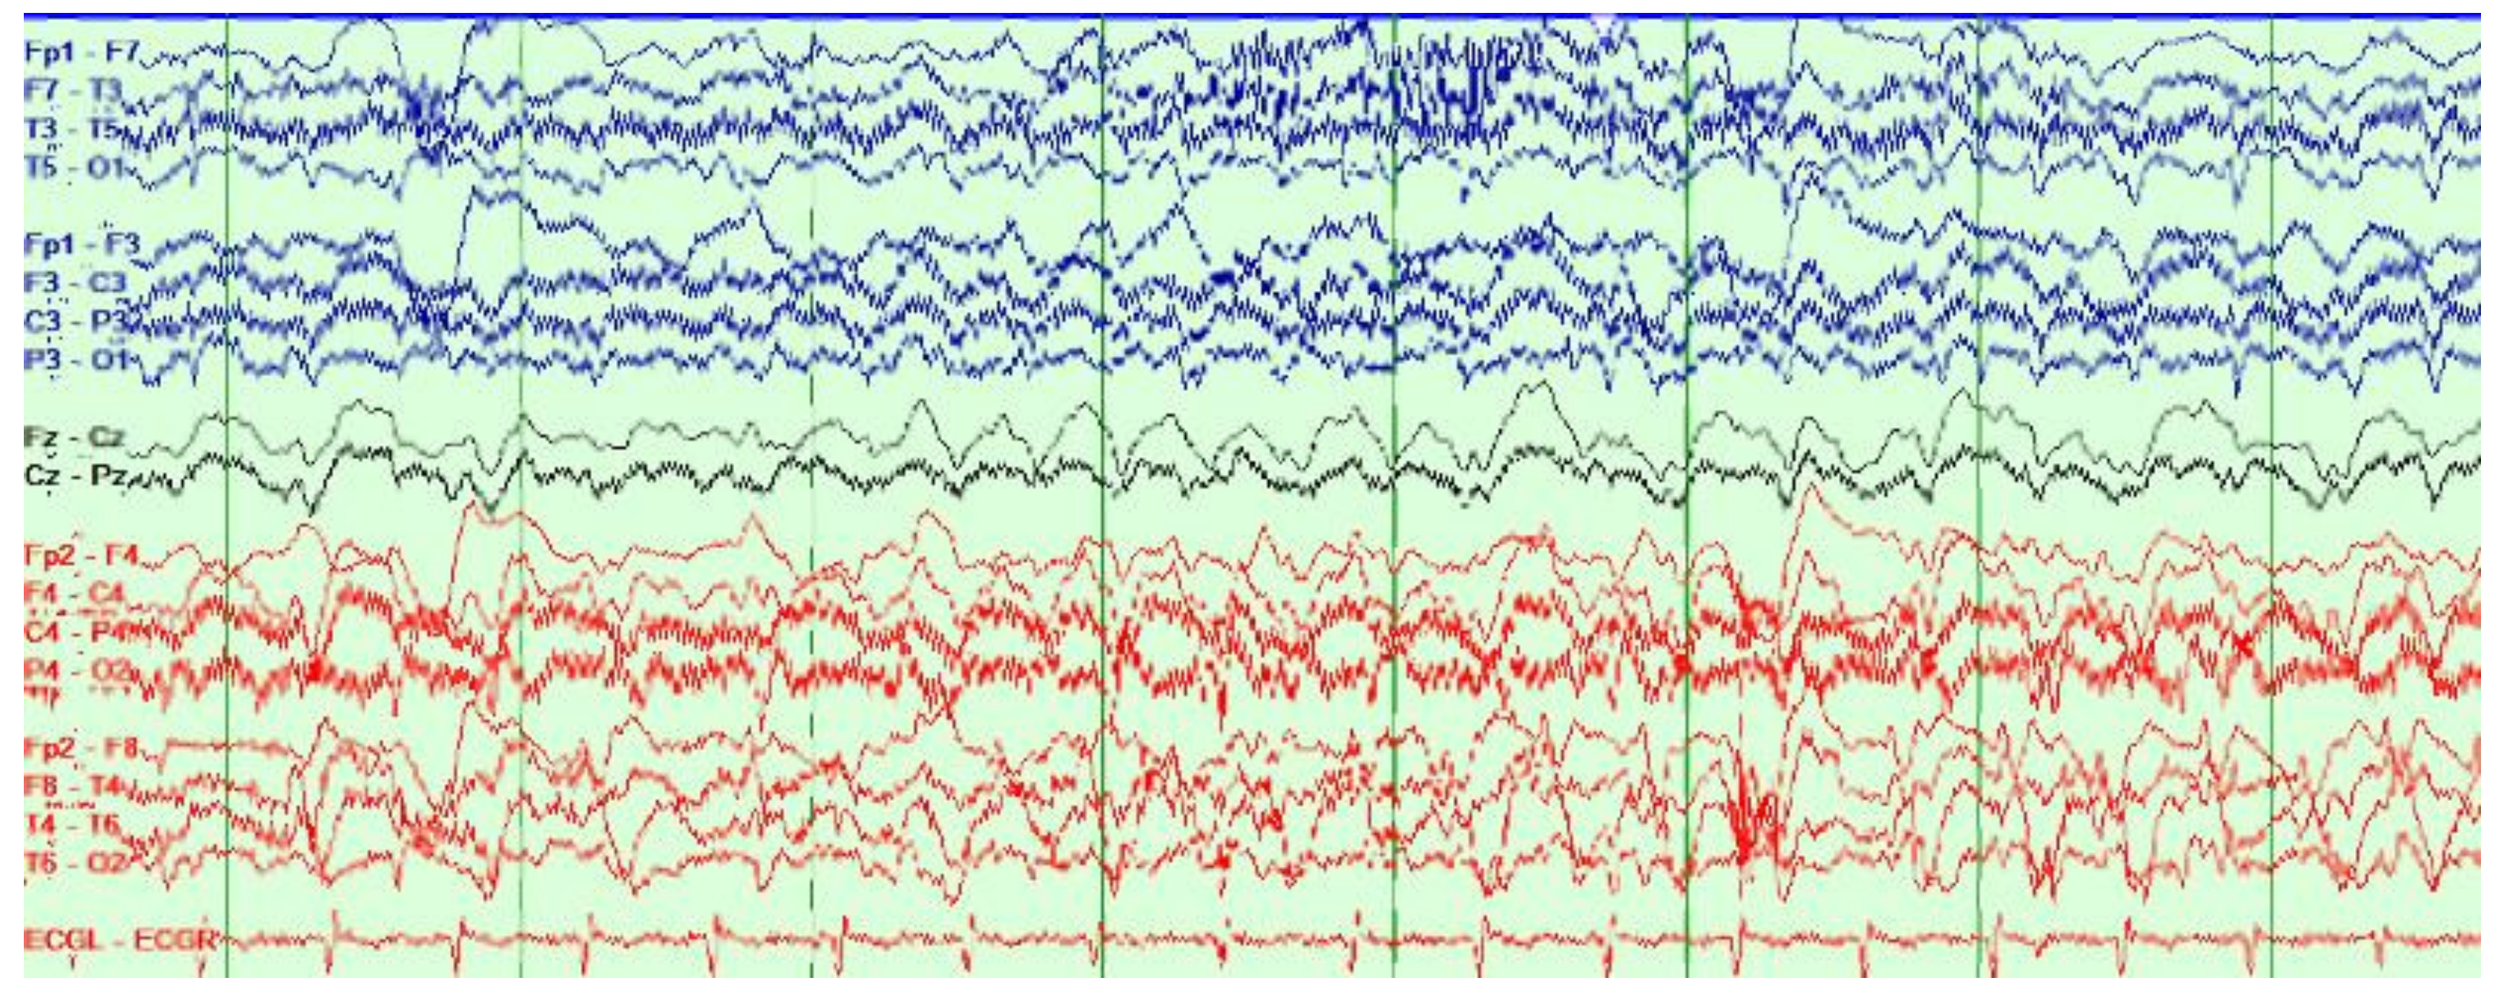
Fp1 - F7
F7 - T3
T3 - T5
T5 - O1
Fp1 - F3
F3 - C3
C3 - P3
P3 - O1
Fz - Cz
Cz - Pz
Fp2 - F4
F4 - C4
C4 - P4
P4 - O2
Fp2 - F8
F8 - T4
T4 - T6
T6 - O2
ECGL - ECGR

This EEG demonstrates the return of electroclinical seizure arising from right centrotemporal region on day 1, about 4 hours after pentobarbital drip was discontinued and as the study drug began. Longitudinal bipolar montage: LFF 1 Hz, HFF 70 Hz; sensitivity, 15 µV/mm; time base, 30 mm/s. EEG, electroencephalogram; HFF, high-frequency filter; LFF, low-frequency filter.

Figure 4B. Double Banana Bipolar Montage—Day 3

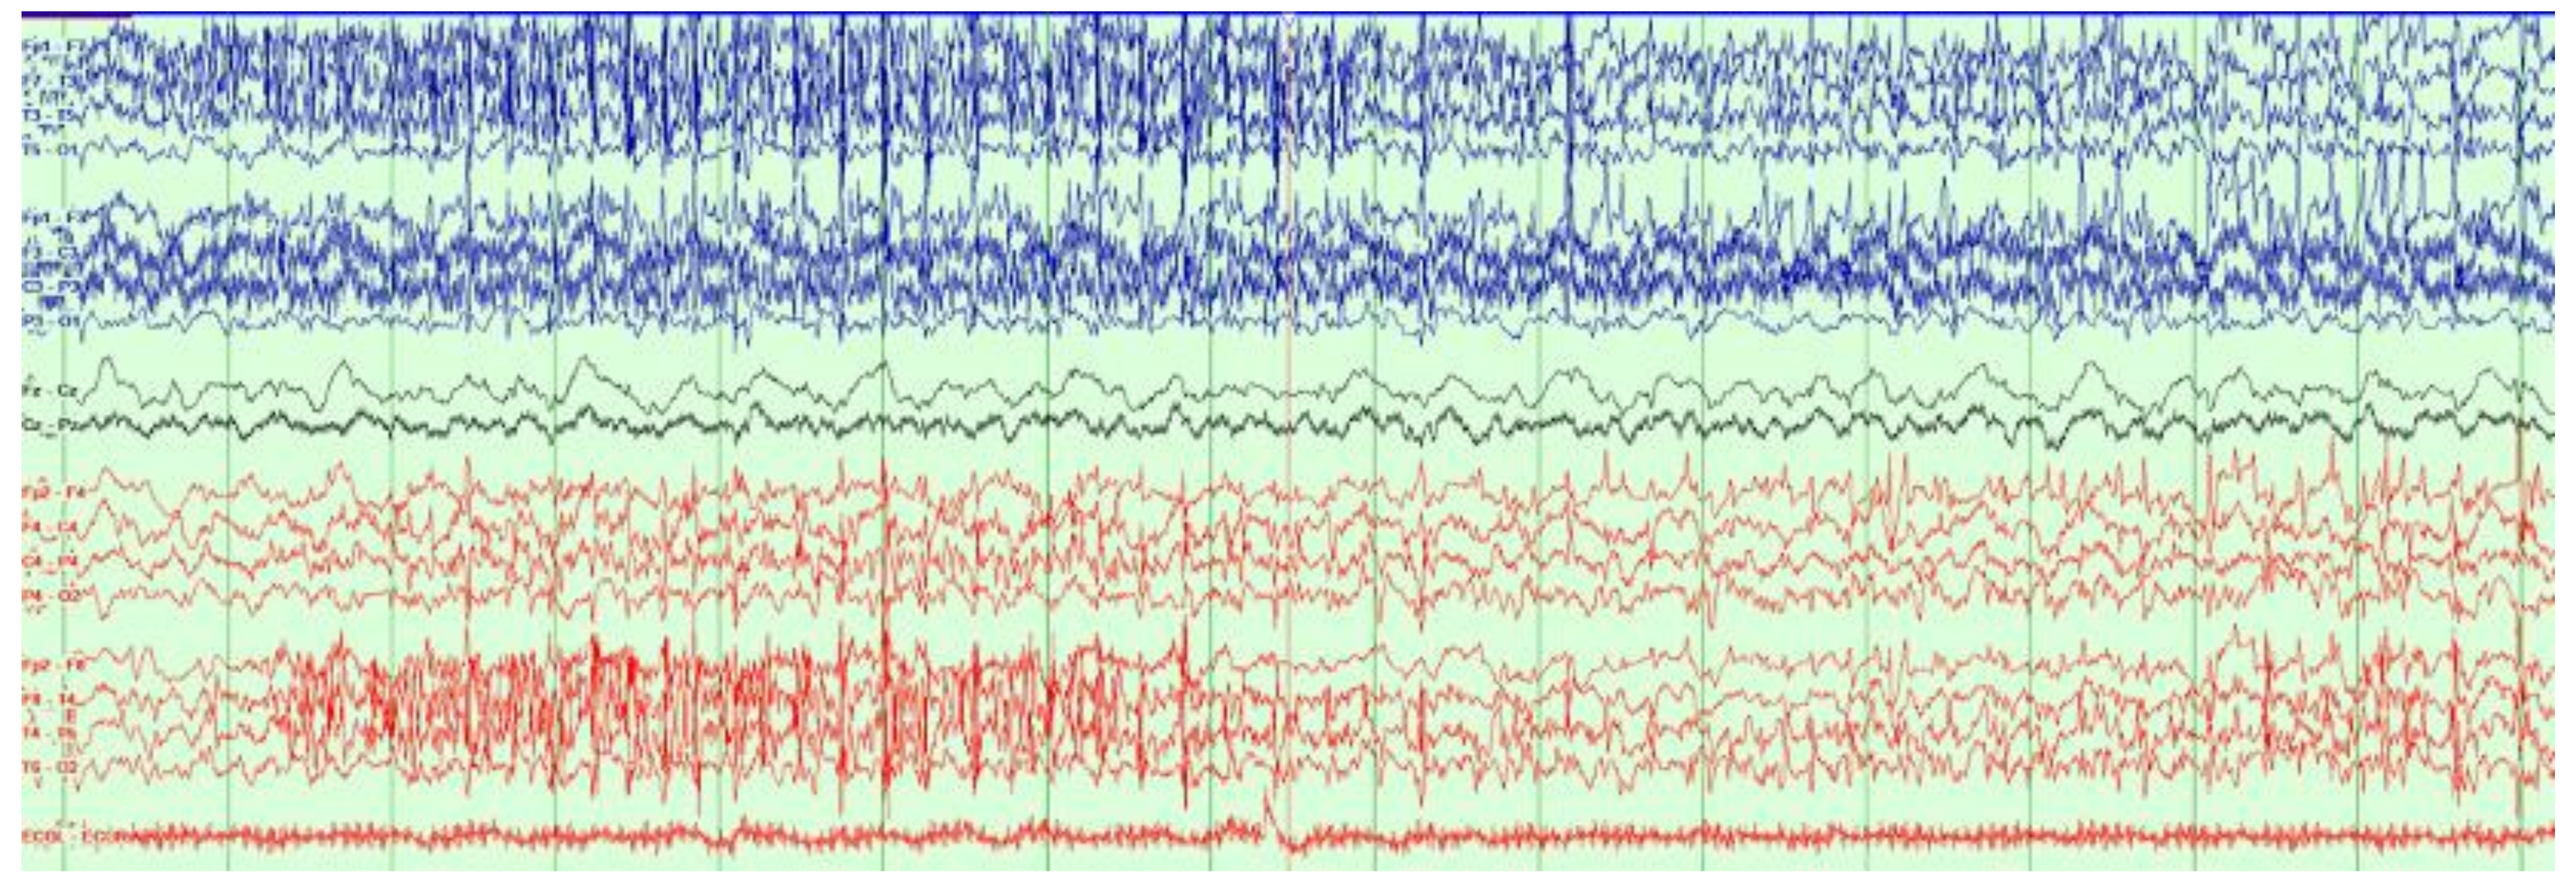

This EEG demonstrates isolated electroclinical seizure into day 3 of protocol as rhythmic fast polyspikes arising from bifrontal regions, which spread anteriorly over both hemispheres. Double banana bipolar montage: LFF 1 Hz, HFF 70 Hz; sensitivity, 15 µV/mm; time base 30 mm/s. EEG, electroencephalogram; HFF, high-frequency filter; LFF, low-frequency filter.

Figure 4C. Double Banana Bipolar Montage—Day 5

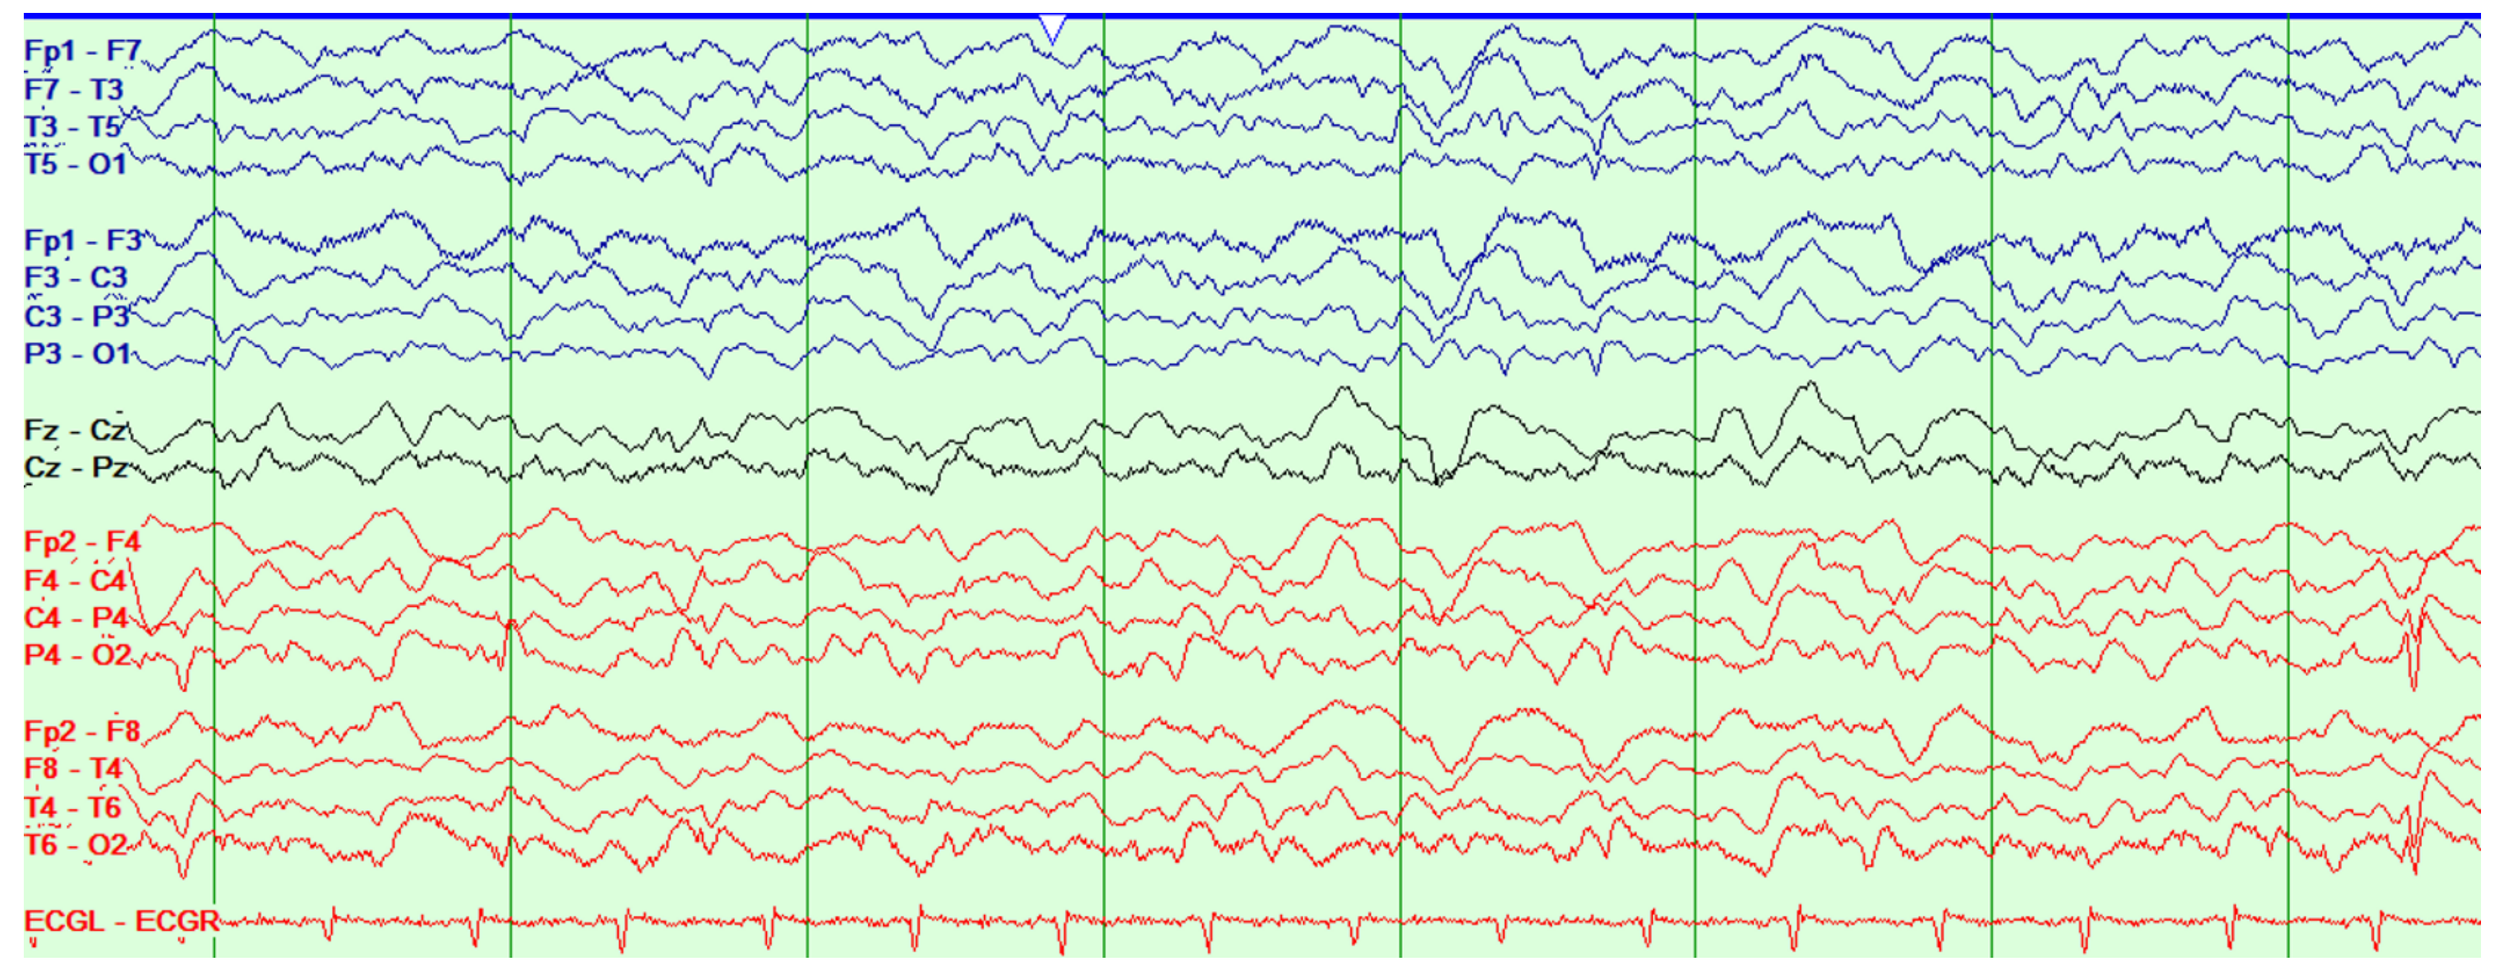
Fp1 - F7
F7 - T3
T3 - T5
T5 - O1
Fp1 - F3
F3 - C3
C3 - P3
P3 - O1
Fz - Cz
Cz - Pz
Fp2 - F4
F4 - C4
C4 - P4
P4 - O2
Fp2 - F8
F8 - T4
T4 - T6
T6 - O2
ECGL - ECGR

This EEG demonstrates cessation of seizures on day 5 of protocol when transitioned from IV to enteric GNX with resolution of seizures, improvement in interictal background, and interictal right occipital epileptiform discharges. Double banana bipolar montage: LFF 1 Hz, HFF 70 Hz; sensitivity, 15 µV/mm; time base, 30 mm/s. EEG, electroencephalogram; HFF, high-frequency filter; LFF, low-frequency filter.

Conclusions

- **Outcomes are generally poor in SRSE**
- **IV GNX is in phase 3 clinical development as a potential treatment for refractory SE**
- **In this patient, GNX was effective in aborting SRSE, allowing for patient improvement and hospital discharge**
- **Additional evaluation of GNX's effect in SRSE is warranted**

References

1. Shorvon S, et al. *Brain*. 2011;134(Pt 10):2802-2818.
2. Sahin M, et al. *Epilepsia*. 2001;42(11):1461-1467.
3. Gilbert DL, et al. *J Child Neurol*. 1999;14(9):602-609.
4. Kravljanac R, et al. *Eur J Paediatr Neurol*. 2015;19(5):584-590.
5. Vasquez A, et al. *Seizures*. 2019;68:62-71.
6. Naylor DE, et al. *J Neurosci*. 2005;25(34):7724-7733.
7. Macdonald RL, et al. *Epilepsia*. 1999;40(suppl 1)S9-S20; discussion S21-S22.
8. Goodkin HP, et al. *J Neurosci*. 2008;28(10):2527-2538.
9. Reddy DS, et al. In: Noebels JL, et al, eds; *Jasper's Basic Mechanisms of the Epilepsies*. 4th ed. National Center for Biotechnology Information; 2012:984-1002.
10. Sullivan J, et al. AES. 2018. Abstract 2.251.

Acknowledgments

Marinus Pharmaceuticals, Inc., provided IV and oral GNX. No monetary support was provided.

Atrium Health Levine Children's

American Epilepsy Society | Virtual | December 4-8, 2020