2.205

Aggregated Safety and Tolerability Experience from the Ganaxolone Development Program

Maciej Gasior, MD, PhD[1]; Joseph Hulihan[1], MD; Ian Miller, MD[1]
[1]*Marinus Pharmaceuticals, Inc., Radnor, PA, USA*

Background

- Ganaxolone (3α-hydroxy-3β-methyl-5α-pregnan-20-one), a neuroactive steroid, is an antiseizure medication (ASM) currently being investigated in patients with genetic epilepsy conditions, including cyclin-dependent kinase-like 5 (CDKL5) deficiency disorder (CDD) and tuberous sclerosis complex (oral formulation), and in the acute setting to treat refractory status epilepticus (intravenous formulation)[1]
 - A new drug application was submitted to the FDA in July 2021 for ganaxolone in the treatment of seizures associated with CDD
- Ganaxolone acts as a positive allosteric modulator of both synaptic and extrasynaptic $GABA_A$ receptors (**Figure 1**). Its broad antiseizure activities were established in multiple preclinical models, which demonstrated that ganaxolone blocks seizure propagation, raises seizure threshold, and can reverse seizures, even with delayed administration.[1]
- To date, the ganaxolone clinical program comprises 24 Phase 1 studies in healthy patients, one Phase 1 study in adults with migraine, and 22 Phase 2/3 studies in adults with epilepsy, infants and children with seizure disorders, children with Fragile X syndrome, adults with post-traumatic stress disorder, women with postpartum depression, adolescents and adults with status epilepticus, and pediatric and adult patients with seizures associated with tuberous sclerosis complex
- A recent, well-controlled Phase 3 study demonstrated statistically significant reductions in major motor seizure frequency in children and young adults with genetically confirmed CDD who were treated with ganaxolone as adjunctive therapy to their standard ASM regimen for up to 17 weeks[2]
- This analysis represents the aggregated safety and tolerability results from all completed, company-sponsored, placebo-controlled, clinical trials in epilepsy and other neuropsychiatric disorders through April 27, 2021

Figure 1. Ganaxolone Is a Positive Allosteric Modulator of Both Synaptic and Extrasynaptic $GABA_A$ Receptors

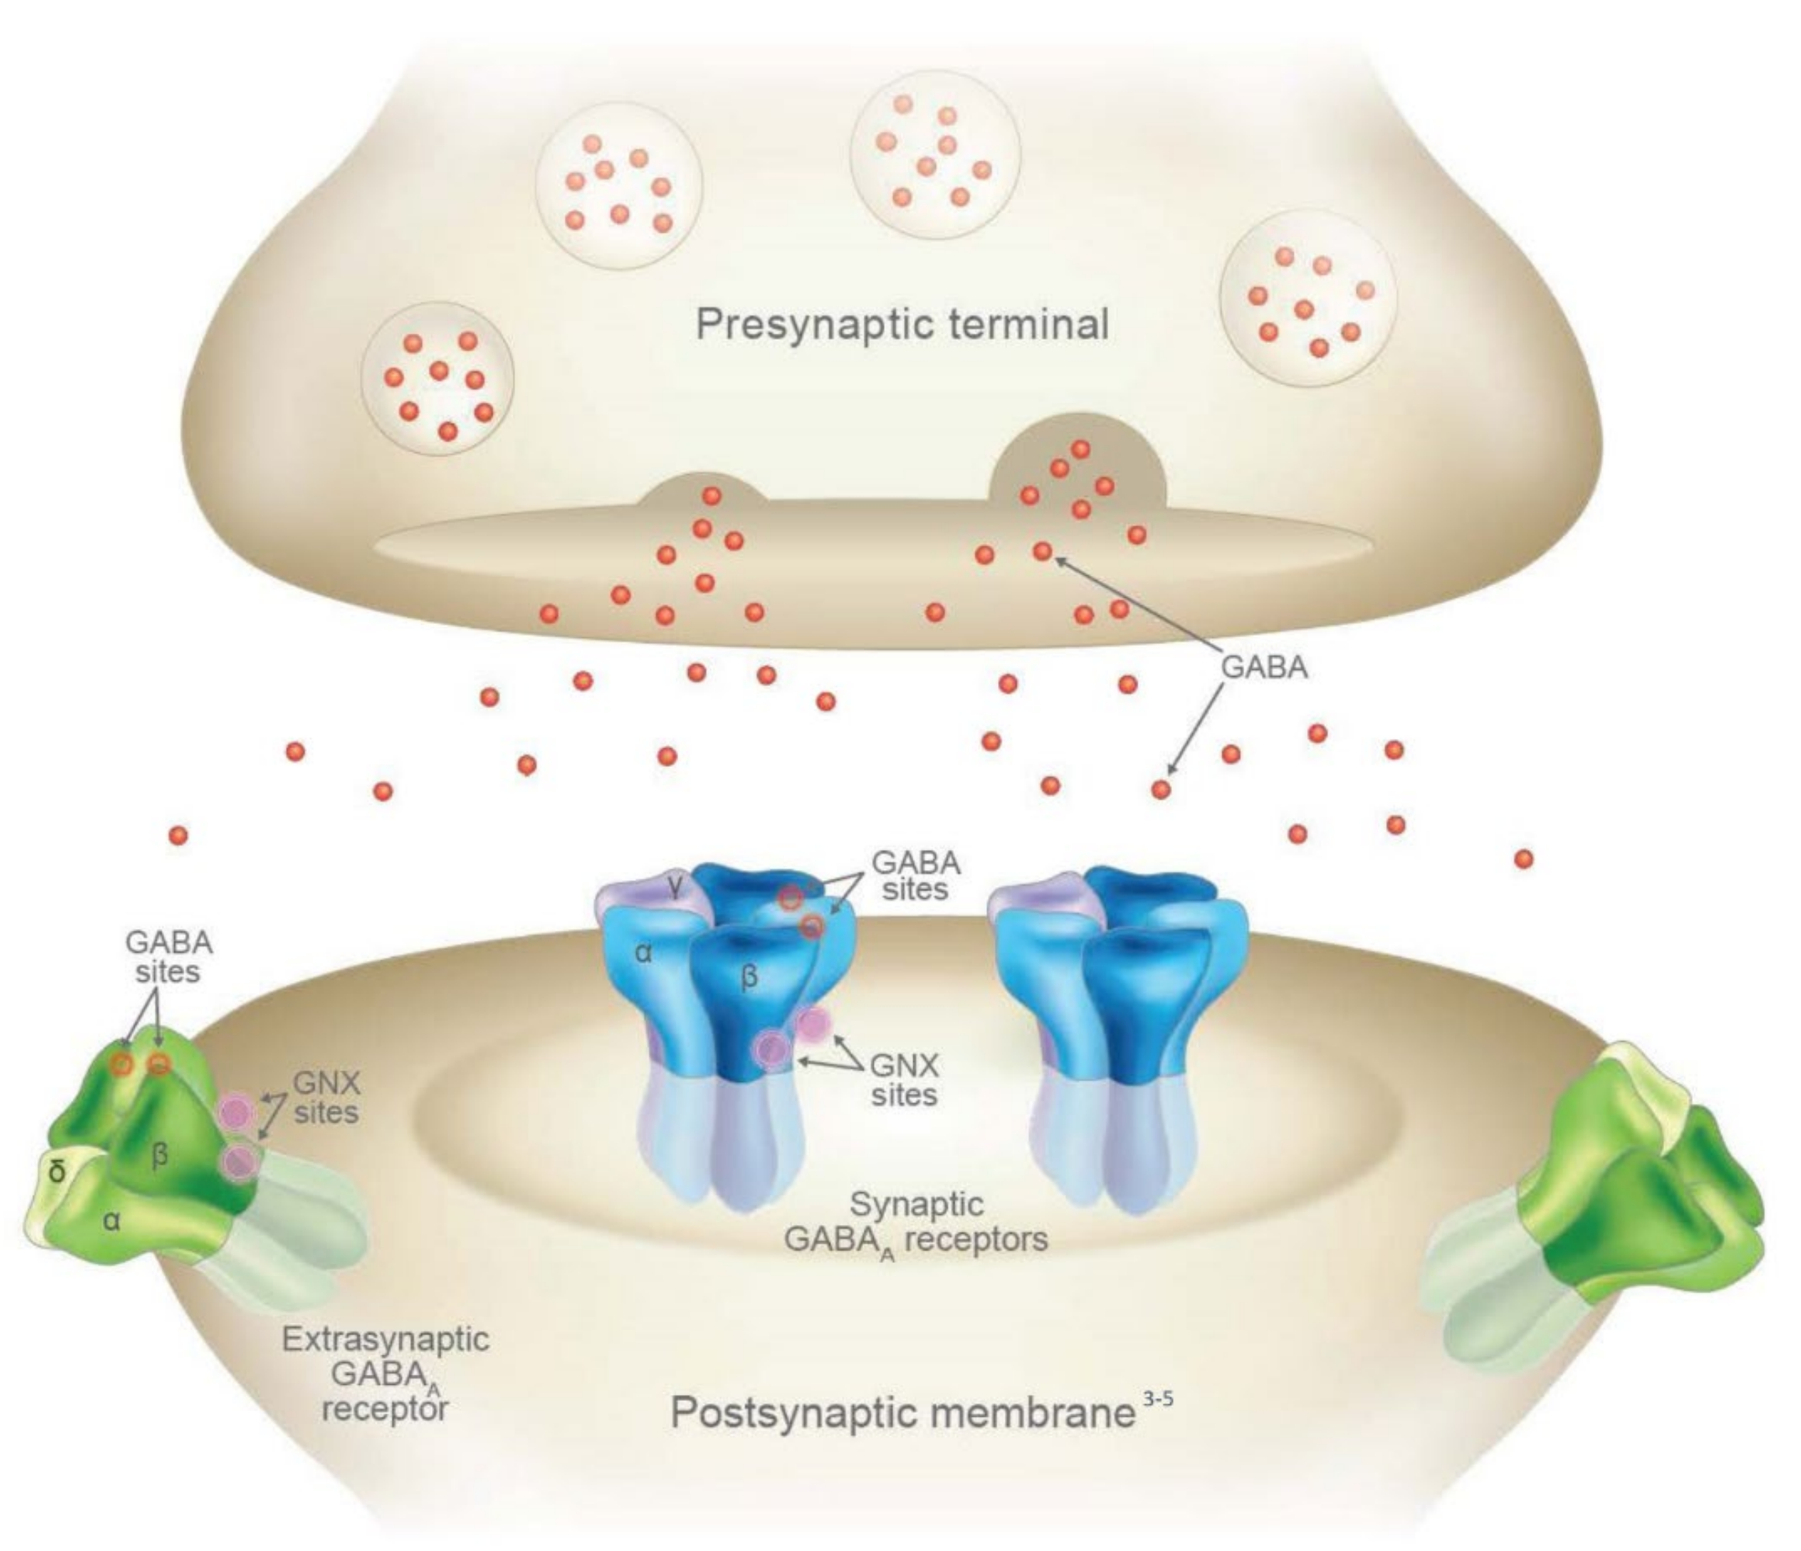
Presynaptic terminal
GABA
GABA sites
GNX sites
GABA sites
GNX sites
Synaptic GABAA receptors
Extrasynaptic GABAA receptor
Postsynaptic membrane 3-5

GNX, ganaxolone.

Methods

Study Design and Patient Population

- Twenty-four Phase 1 and 22 Phase 2/3 company-sponsored, placebo-controlled studies were included in this aggregated summary of safety and tolerability
 - All patients received at least 1 dose of either ganaxolone or placebo
 - Age ranged from 4 months to 88 years

Analyses

- Safety assessments included the incidence of treatment-emergent adverse events (TEAEs), monitoring of vital signs, physical examinations, electrocardiograms (ECGs), and laboratory tests
 - The frequency, severity, and MedDRA classification of AEs, and their relation to treatment were analyzed
 - In addition to the AEs that occurred during the treatment period and during the follow-up period of the clinical studies, AEs that were present at pretreatment and worsened in severity during the trials were included in the summary and integrated by study phase, formulation, and indication
 - Potential adverse drug reactions (ADRs) were analyzed

Results

Patient Characteristics and Baseline Demographics

- 1844 patients were included in the analysis; 1101 received ganaxolone and 743 received placebo
 - More than 75% of patients included in the analysis were from Phase 2 or 3 studies

Treatment-emergent Adverse Events

- TEAEs were reported in 62.9% (693/1101) patients who received ganaxolone and 53.8% (400/743) patients who received placebo (**Table 1**)
 - The most frequently reported TEAEs (≥5% of patients) in ganaxolone-treated patients were somnolence, dizziness, fatigue, and headache
 - All but headache occurred more frequently in the ganaxolone-treated patients

Table 1. Treatment-emergent Adverse Events in Completed Placebo-controlled Studies Reported by ≥5% of patients

	Ganaxolone (n=1101)	Placebo (n=743)
Total number of patients with at least one AE, n (%)		
	693 (62.9)	400 (53.8)
TEAEs ≥5% by SOC Preferred Term, n (%)		
Somnolence	247 (22.4)	60 (8.1)
Dizziness	139 (12.6)	29 (3.9)
Headache	60 (5.4)	52 (7.0)
Fatigue	102 (9.3)	36 (4.8)

AE, adverse event, SOC, system organ class.

- CNS-related events appeared to be dose-related, with the majority of events occurring at ganaxolone doses ≥500 mg
- There were no TEAEs reported by >5% of study participants in the ganaxolone group that also occurred with greater frequency than in the placebo-treated group for the following system organ classes and preferred terms: Gastrointestinal Disorders (vomiting [3.8%], nausea [2.4%], diarrhea [2.3%], constipation [1.2%]); Infections and Infestations (upper respiratory tract infection [2.7%]), pneumonia bacteria/mycoplasmal [0]); Psychiatric Disorders (suicidal ideation [0.1]/attempt 0, insomnia [1.2%], agitation [0.7%]); Skin and Subcutaneous Disorders (rash [1.8%]); Investigations (hepatic enzyme increased [0.2 %], liver function test abnormal [0.2%], transaminases increased [0.2%], ammonia increased [0.1%])

Discontinuation Adverse Events (DAEs)

- Among TEAEs that led to discontinuation of ≥2 patients, CNS-related events, rash, constipation, and hypertension were reported more frequently by patients in the ganaxolone group compared to those in the placebo group
 - The most common CNS-related AEs that led to discontinuation were dizziness (ganaxolone 1.1%, placebo 0.4%), somnolence (0.9%, placebo 0.4%), fatigue (ganaxolone 0.8%, placebo 0.1%), and gait disturbance (ganaxolone 0.1%, placebo 0%)

Severity, Serious Adverse Events, and Deaths

- The majority of TEAEs were nonserious and mild to moderate in severity
- The frequency of serious adverse events (SAEs) was similar between ganaxolone- and placebo-treated patients: 2.8% (31/1101) and 3.8% (28/743), respectively
 - Seizure was the most common SAE and was reported by 0.5% (5/1101) of patients in the ganaxolone treatment group and 0.7% (5/743) patients in the placebo group
- SAEs that were considered treatment related by either the investigator or the sponsor were rare: 1.6% (18/1101) of the patients who received ganaxolone and 1.7% (13/743) of the placebo-treated group (**Figure 2**)

Figure 2. Most Common SAEs Considered to be Treatment-related by the Investigator and/or the Sponsor

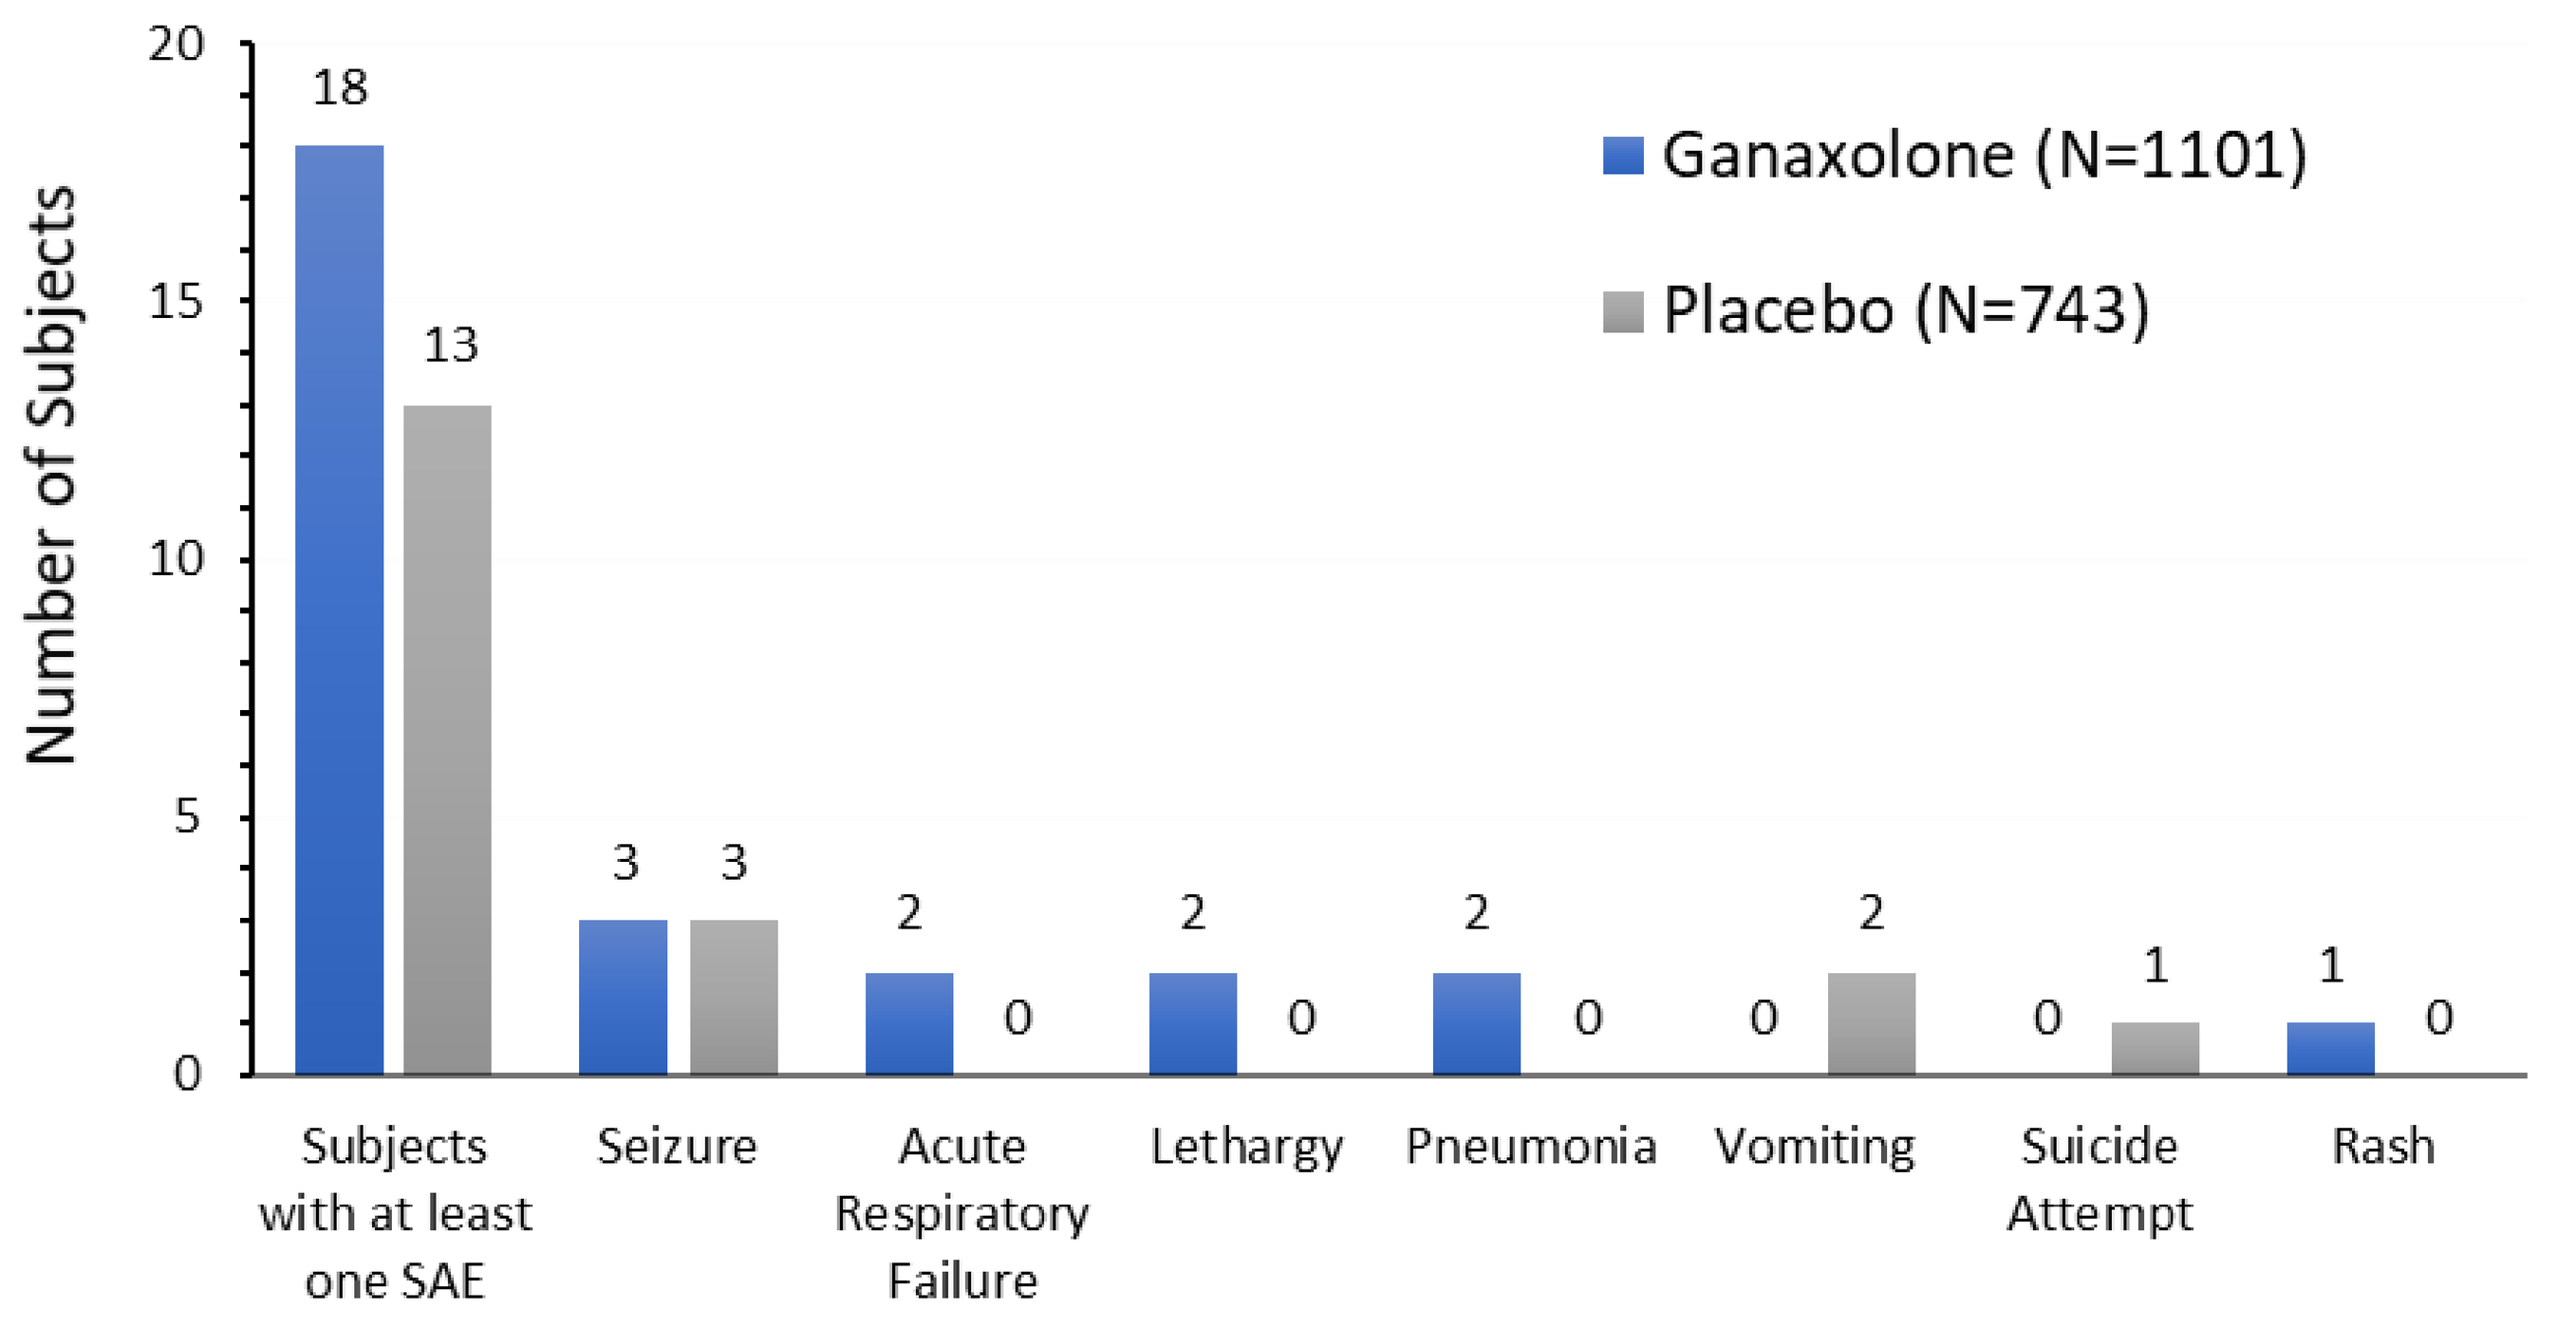
Number of Subjects
20
15
10
5
0
18
13
3
3
2
0
2
0
2
0
0
2
0
1
1
0
Subjects with at least one SAE
Seizure
Acute Respiratory Failure
Lethargy
Pneumonia
Vomiting
Suicide Attempt
Rash
Ganaxolone (N=1101)
Placebo (N=743)

SAE, serious adverse event.

- Seven deaths (6 in patients who were treated with ganaxolone) occurred; none of which were assessed by the investigator as being related to the study drug

Adverse Drug Reactions (ADRs)

- TEAEs considered to be potential adverse drug reactions (ADRs) are summarized in **Table 2**
- Potential ADRs included any TEAE with a frequency of ≥2% in the ganaxolone-treatment group and reported at 2x the frequency of placebo
- All events considered potential ADRs were CNS related; this finding is anticipated based on the mechanism of action of ganaxolone and is consistent with the findings in the non-clinical studies

Table 2. Summary of Potential ADRs

Preferred Term	Ganaxolone (n=1101) n (%)	Placebo (n=743) n (%)
Somnolence	247 (22.4)	60 (8.1)
Dizziness	139 (12.6)	29 (3.9)
Asthenia	27 (2.5)	6 (0.8)
Gait disturbance	26 (2.4)	8 (1.1)
Coordination abnormal	23 (2.1)	6 (0.8)

ADR, adverse drug reaction; Adverse events are classified according to MedDRA version 23.0.

- No discernable safety signals related to bone marrow suppression, bone mineralization, nephrolithiasis, cardiac valvulopathy, or liver function were reported
- No significant changes were noted in body weight and no trends in clinically significant changes in ECG parameters or vital signs in patients who received ganaxolone were reported
- For TEAEs across studies, the safety profile from individual Phase 1 studies appeared to be consistent with the findings from the Phase 2/3 studies

Conclusions

- The aggregated safety and tolerability experience with ganaxolone in clinical trials suggests a consistent and acceptable tolerability and safety profile
- The majority of TEAEs were dose-dependent, non-serious, and mild to moderate in severity
- The most common TEAEs were related to the CNS and anticipated based on the mechanism of action of ganaxolone

References

1. Nohria V, Giller E. *Neurotherapeutics.* 2007;4:102-5.
2. Pestana-Knight E, et al. [poster]. Presented at American Epilepsy Society, Virtual, December 4-8, 2020; 2020.

Acknowledgments

This study was supported by Marinus Pharmaceuticals, Inc. (Radnor, PA). Medical writing assistance was provided by Susan Manganaro, MD, MBA, of MedVal Scientific Information Services, LLC (Princeton, NJ) and was funded by Marinus Pharmaceuticals, Inc.

Disclosures

MG: is an employee and shareholder of Marinus Pharmaceuticals, Inc.
JH: is an employee and shareholder of Marinus Pharmaceuticals, Inc.
IM: is an employee and shareholder of Marinus Pharmaceuticals, Inc.

Presented at the American Epilepsy Society Annual Meeting, December 3-7, 2021, Chicago, IL.