Ganaxolone Administration Via G-tube: Subgroup Analysis of the Phase 3 Marigold Study in CDKL5 Deficiency Disorder (CDD)

Maciej Gasior, MD, Alex Aimetti, PhD, Joe Hulihan, MD, Lisa Alvarez, PharmD

Marinus Pharmaceuticals, Inc., Radnor, PA

Background

- Ganaxolone is a neuroactive steroid and positive allosteric modulator that targets both synaptic and extrasynaptic $GABA_A$ receptors
- Ganaxolone is approved in the United States for the treatment of seizures associated with CDKL5 Deficiency Disorder (CDD) in patients ≥2 years old
- CDD presents with treatment-refractory seizures, severe global developmental impairment, and multiple comorbidities[1]
- Ganaxolone is administered as an oral suspension, however ~20% of patients with CDD are fed exclusively by gastrostomy (g-tube) or nasogastric tubes[2]
- The Marigold study was a global, phase 3, randomized, double-blind, placebo-controlled study of patients with CDD treated with adjunctive ganaxolone

Objective

- Analyze the safety and efficacy of the subgroup of patients with feeding tubes in the Marigold study

Methods

- Marigold Study Design
 - A total of 101 patients with CDD aged 2-19 years were enrolled in the study
 - 6-week prospective baseline period followed by a 17-week double-blind treatment phase (**Figure 1**)
 - Seizure data were collected at baseline and at 17 weeks of ganaxolone treatment
 - Primary endpoint was percent change in 28-day major motor seizure frequency (MMSF) during the 17-week treatment phase in relation to the 6-week baseline
 - Major motor seizures were defined as bilateral tonic, bilateral clonic, generalized tonic-clonic, atonic/drop, focal to bilateral tonic-clonic
- 17 patients had a g-tube in place prior to enrollment
 - 10 were randomized to the ganaxolone group
 - 7 were randomized to the placebo group
- Ganaxolone was taken 3 times a day at a maintenance dose of up to 63 mg/kg/day or 1800 mg/day

Figure 1. Marigold Study Design

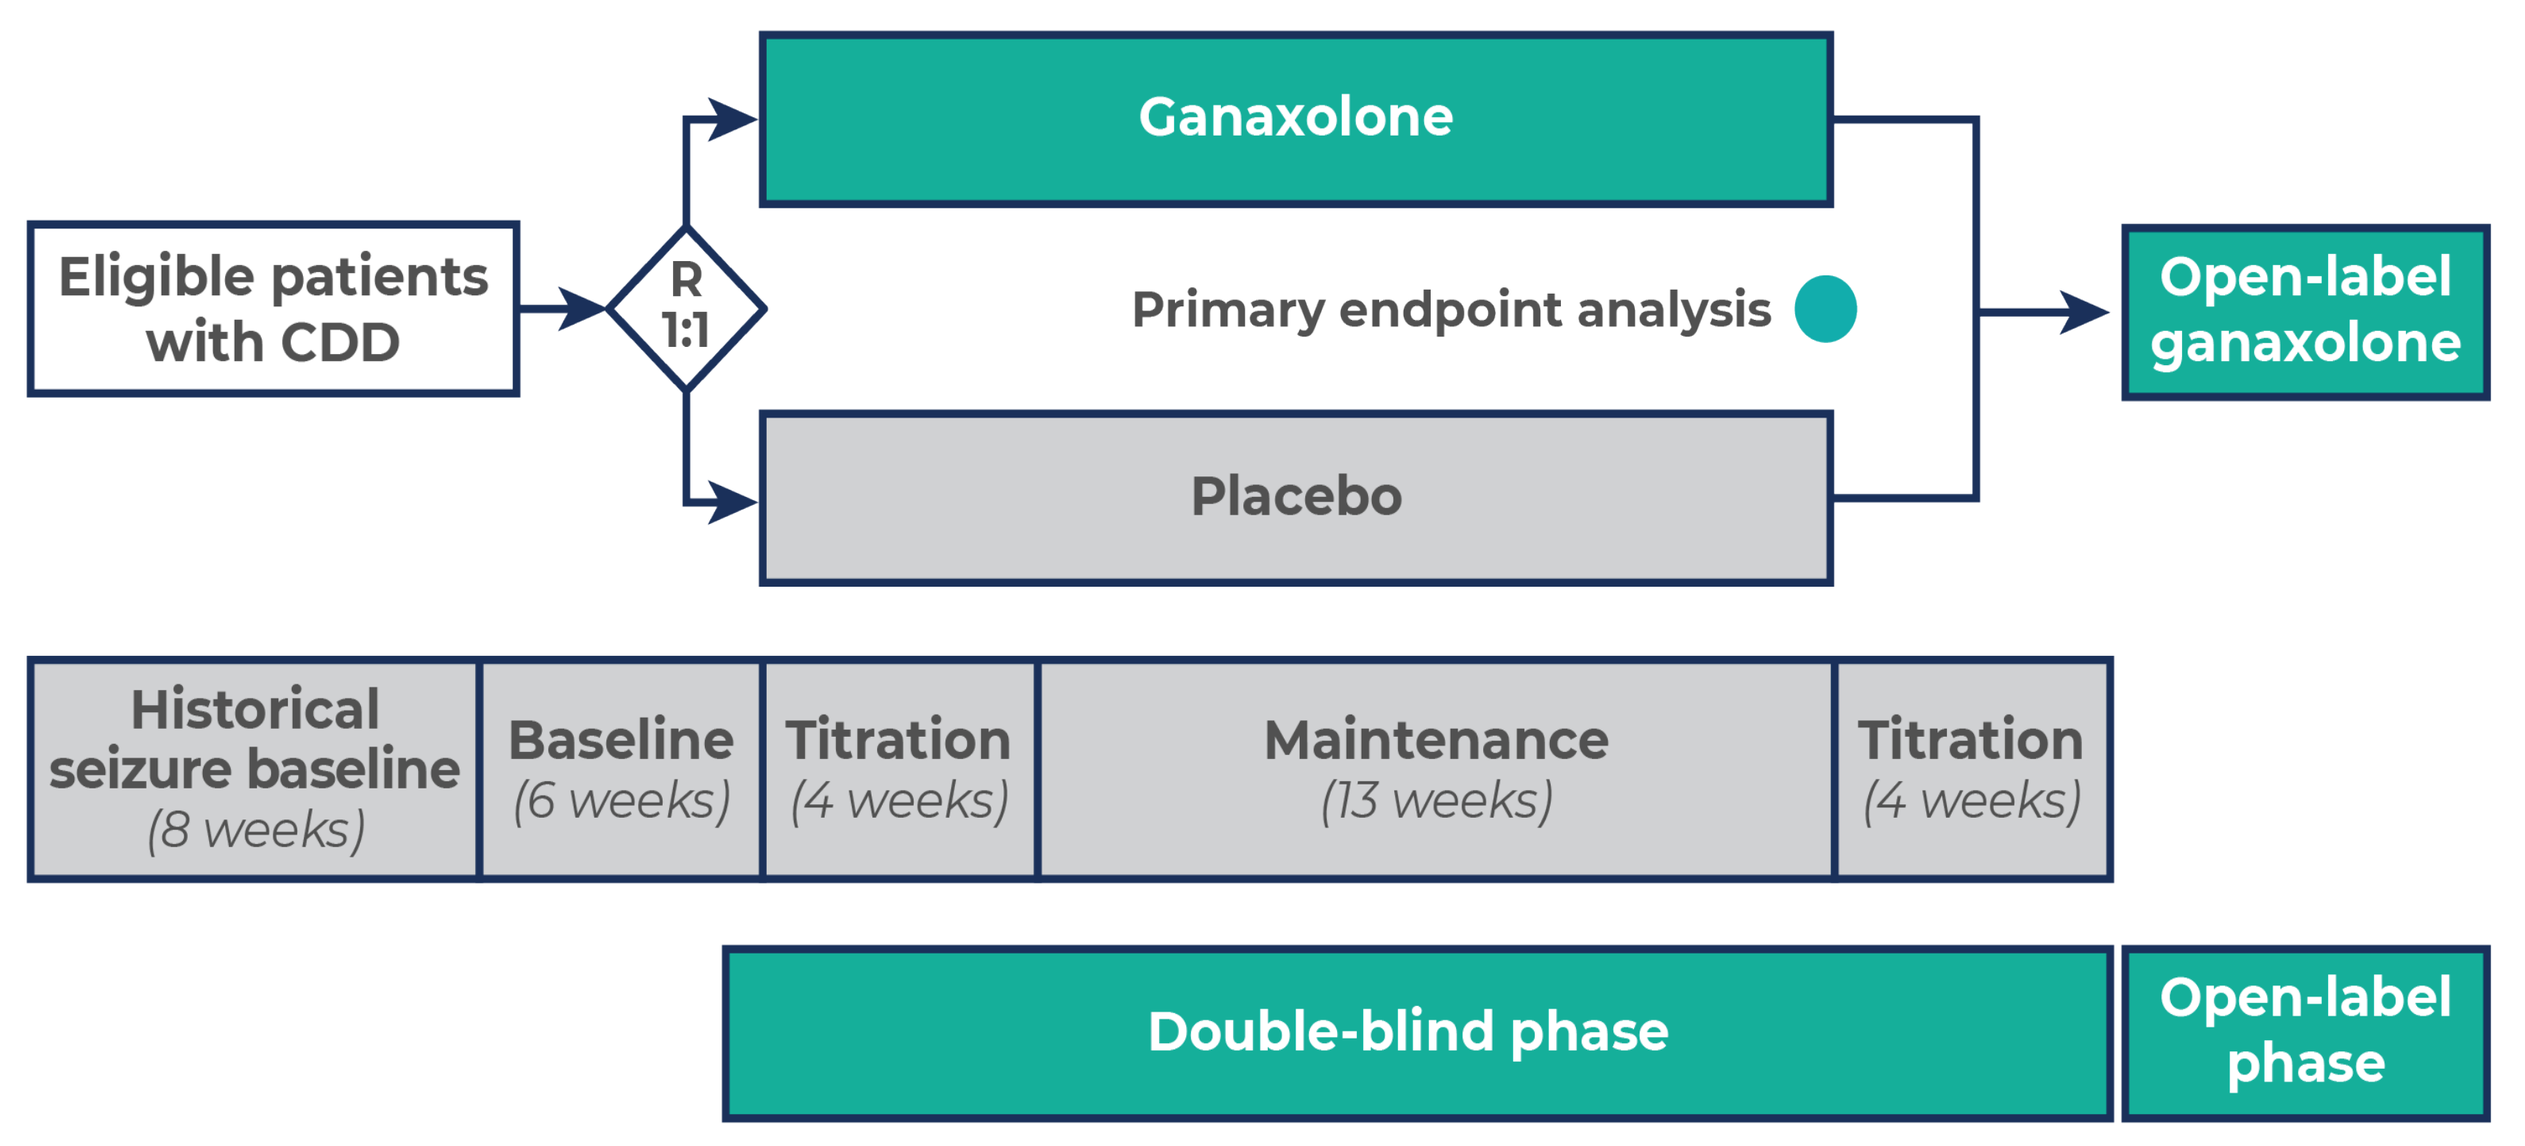
Eligible patients with CDD
R
1:1
Ganaxolone
Primary endpoint analysis
Placebo
Open-label ganaxolone
Historical seizure baseline (8 weeks)
Baseline (6 weeks)
Titration (4 weeks)
Maintenance (13 weeks)
Titration (4 weeks)
Double-blind phase
Open-label phase

CDKL5, cyclin-dependent kinase-like 5; CDD, CDKL5 deficiency disorder; R, randomization.

- Outcomes
 - Percent change in 28-day MMSF
 - Plasma ganaxolone concentrations
 - Treatment-emergent adverse events (TEAEs)
 - TEAE severity

Results

Table 1. Enrollment and Baseline Demographics of Entire Marigold Population by Route of Administration

	G-tube			No G-tube		
	Ganaxolone (n=10)	Placebo (n=7)	Total (n=17)	Ganaxolone (n=40)	Placebo (n=44)	Total (n=84)
Age, yr, median (Q1-Q3)	8 (5, 13)	10 (5-15)	10 (5-13)	4 (3-9)	7 (3.5-10.5)	5 (3-10)
Sex, n (%)						
Male	4 (40)	1 (14)	5 (29)	7 (18)	9 (21)	16 (19)
Female	6 (60)	6 (86)	12 (71)	33 (83)	35 (80)	68 (81)
Ethnicity, n (%)						
Hispanic or Latino	1 (10)	0	1 (6)	3 (8)	6 (14)	9 (11)
Not Hispanic or Latino	9 (90)	7 (100)	16 (94)	35 (88)	36 (83)	71 (85)
Unknown	0	0	0	1 (3)	1 (2)	2 (2)
Not reported	0	0	0	1 (3)	1 (2)	2 (2)
Race, n (%)						
White	9 (90)	7 (100)	16 (94)	37 (93)	40 (91)	77 (92)
Asian	0	0	0	2 (5)	3 (7)	5 (6)
Other	1 (10)	0	1 (6)	1 (3)	1 (2)	2 (2)

Table 2. Baseline Clinical Characteristics of Entire Marigold Population by Route of Administration

	G-tube			No G-tube		
	Ganaxolone (n=10)	Placebo (n=7)	Total (n=17)	Ganaxolone (n=40)	Placebo (n=44)	Total (n=84)
Baseline MMSF per 28 days, median (IQR)	133.3 (48.5-279.1)	60.0 (49.2-84.0)	60.7 (49.2-253.3)	45.5 (30.7-141.3)[a]	43.8 (17.7-125.9)	45.3 (35.3-63.9)[b]
Number of ASMs taken prior to the study, median (IQR)	10 (8-13)[c]	7 (3-11)	9 (5.5-11.5)[d]	7 (5-10)[e]	7 (4-9)[f]	7 (5-9)[g]
Number of concomitant ASMs, median (IQR)	2 (1-3)	3 (2-4)	2 (2-3)	3 (2-4)	2 (1-3)	2 (1.5-3)
Concomitant ASMs[h], n (%)						
Valproate	2 (20)	4 (6)	6 (35)	16 (40)	12 (27)	28 (33)
Levetiracetam	2 (20)	0	2 (12)	11 (28)	13 (30)	24 (29)
Clobazam	4 (40)	4 (6)	8 (47)	8 (20)	10 (23)	18 (21)
Vigabatrin	1 (10)	0	1 (6)	9 (23)	12 (27)	21 (25)

ASM, antiseizure medication. [a]n=39; [b]n=83; [c]n=9; [d]n=16; [e]n=39; [f]n=43; [g]n=82; [h]The 4 most common concomitant ASMs.

Figure 2. Median (95% CI) % Change in MMSF Shows No Difference Between the G-tube and No G-tube Groups

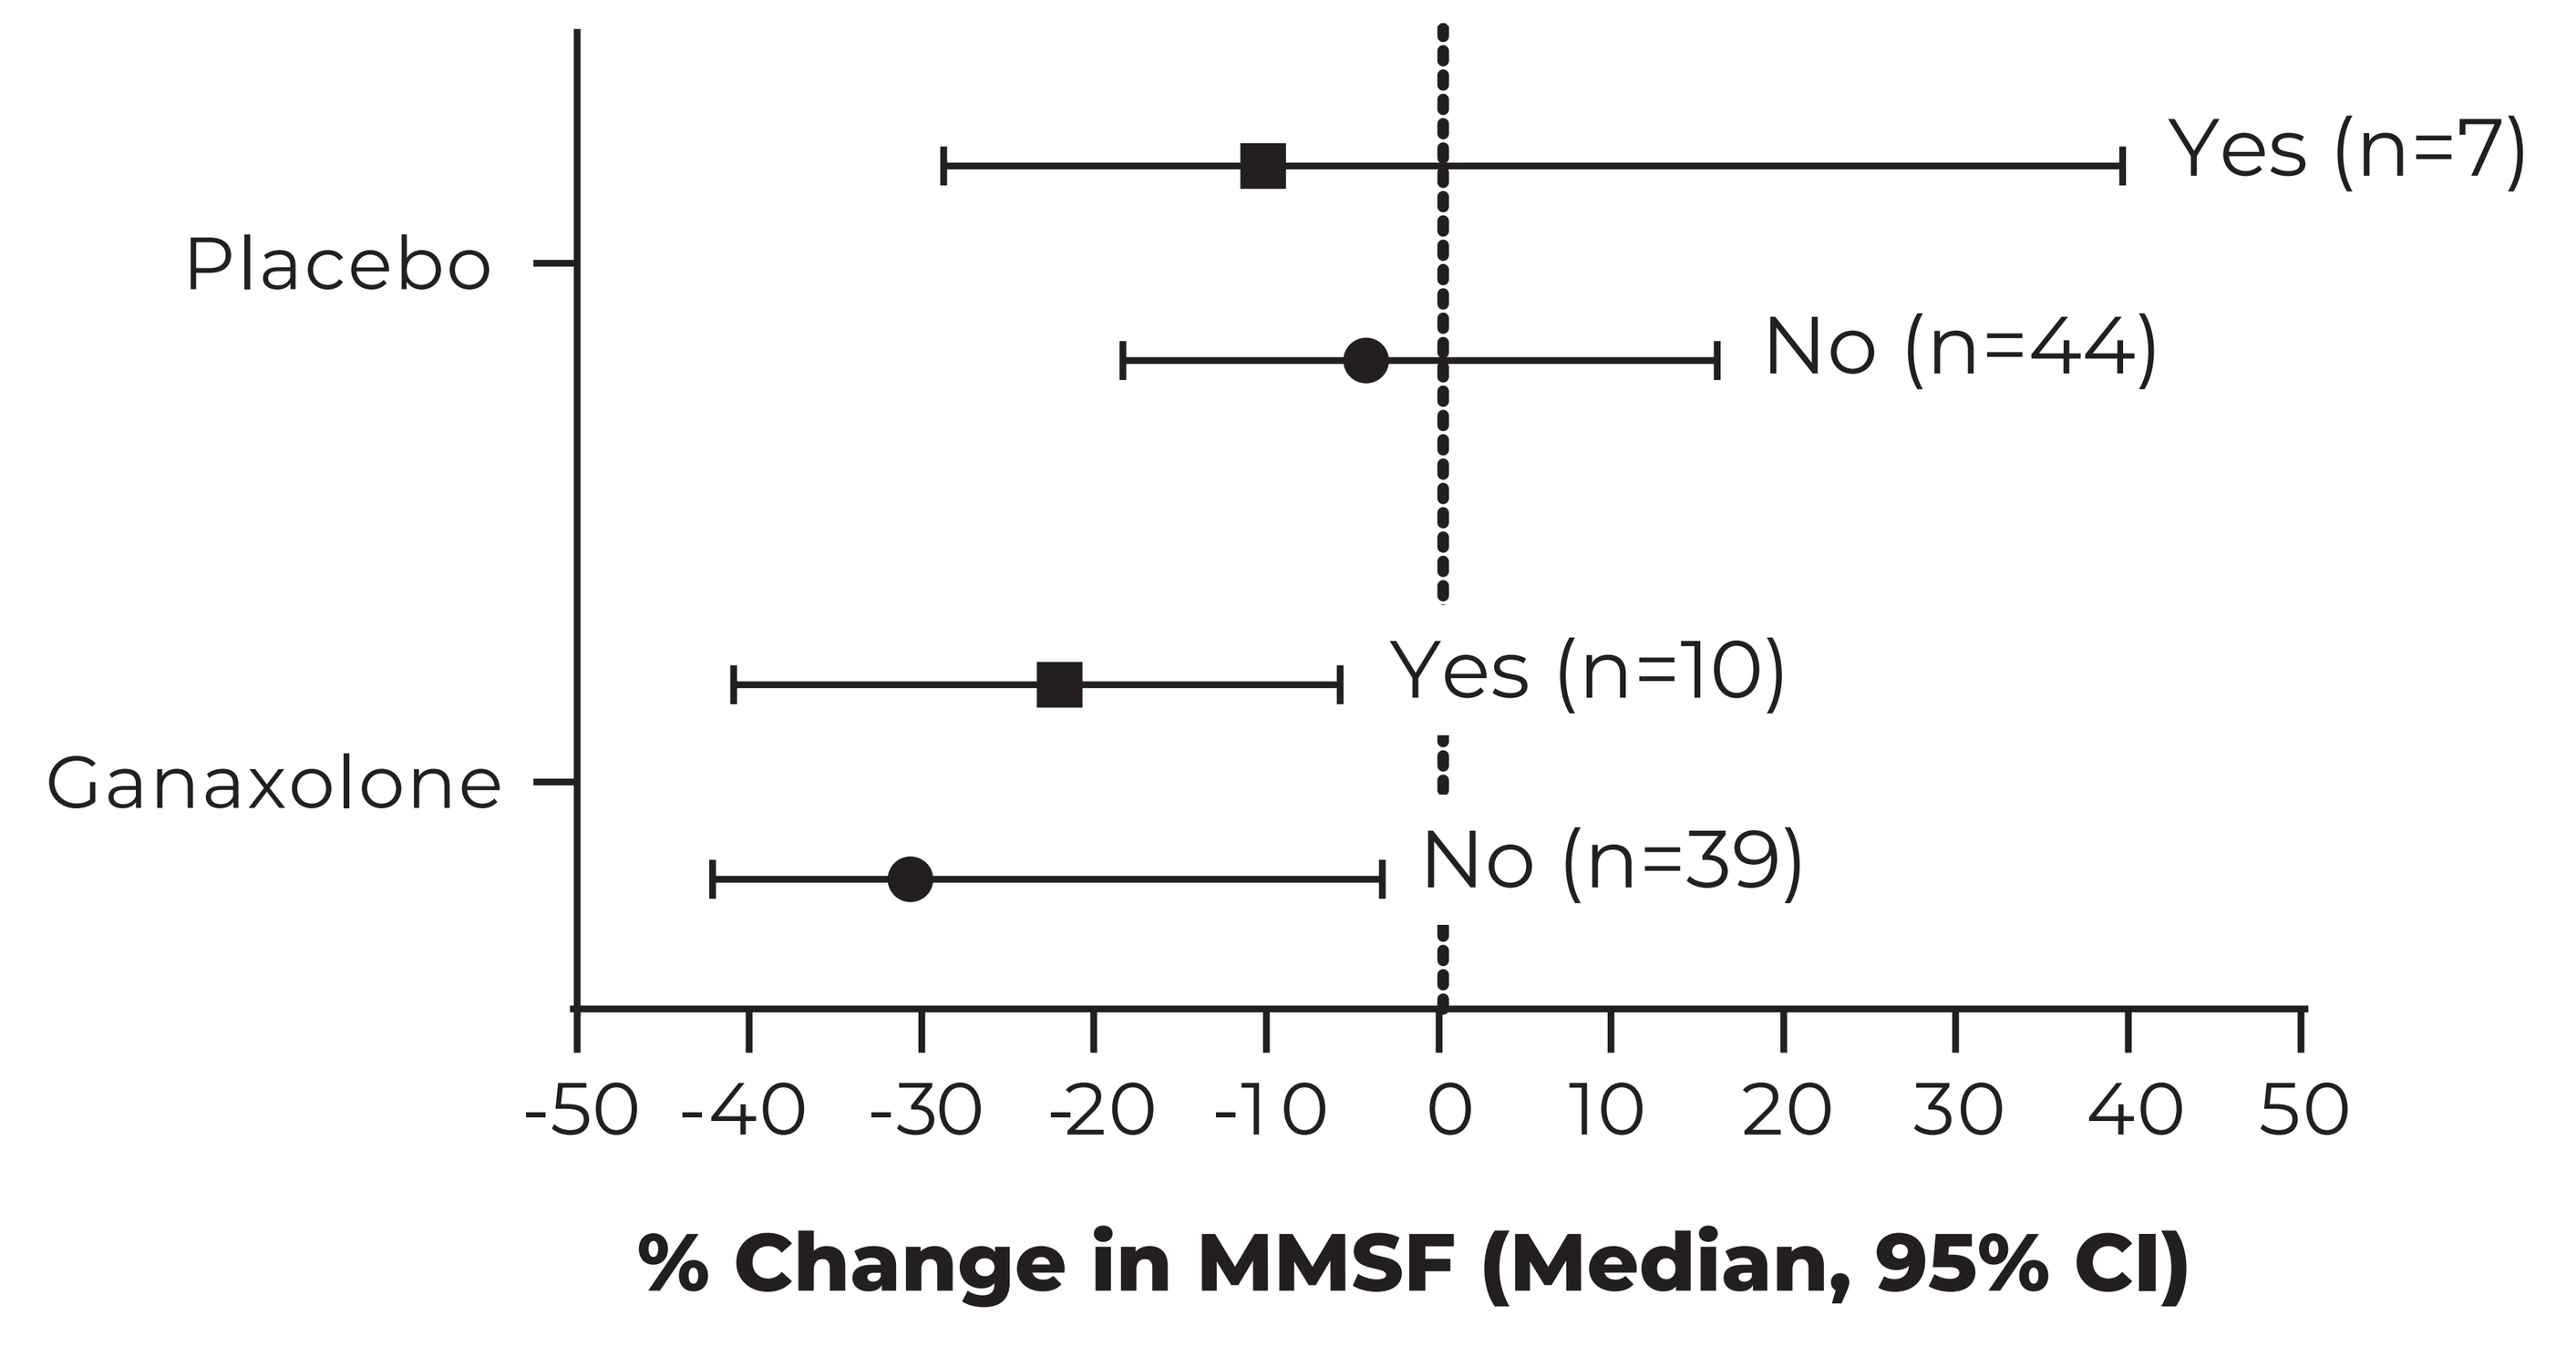
Placebo
Yes (n=7)
No (n=44)
Ganaxolone
Yes (n=10)
No (n=39)
-50 -40 -30 -20 -10 0 10 20 30 40 50
% Change in MMSF (Median, 95% CI)

- Percent change in MMSF was consistent between patients with (yes) and without (no) drug administered via g-tube

Table 3. Summary of Ganaxolone Concentration Level (ng/mL) for Patients With or Without Drug Administered via G-tube, 17-week, Double-Blind Phase, Intent-to-Treat Population

Time Point	Drug via G-tube (number of patients)	Mean (SD)	Median (min, max)
Pre-dose[a]	No (29)	85.93 (75.55)	61.7 (13.2, 348)
	Yes (5)	92.34 (65.22)	64.2 (44.5, 204)
0-4 hr post-dose	No (25)	106.54 (71.68)	82.8 (21.2, 336)
	Yes (6)	92.35 (51.15)	89.4 (34, 181)
1-5 hr post-dose	No (11)	119.7 (107.55)	94.7 (24.7, 325)
	Yes (2)	46.4 (31.67)	46.4 (24, 68.8)
4-8 hr post-dose	No (25)	103.04 (83.99)	61.4 (28.1, 341)
	Yes (7)	90.31 (73.54)	70 (29.9, 246)

[a]Pre-dose concentration levels taken at end of Week 17.

- Trough ganaxolone levels were comparable in patients with and without g-tubes, indicating little potential for compromised efficacy in patients receiving ganaxolone via g-tube. Likewise post-dose plasma ganaxolone levels showed no clinically meaningful differences in the two groups.

Safety

- Adverse events occurred in 80% of patients with g-tubes and 87.5% of patients without g-tubes who received ganaxolone
- Two patients taking ganaxolone were hospitalized due to TEAEs: urinary tract infection of moderate severity and decreased oxygen saturation. The latter was considered related to study drug.
- With or without a g-tube, the type, frequency, and severity of TEAEs were similar
- TEAEs were mild or moderate; there were no severe events

Table 4. Treatment-Emergent Adverse Events in Both G-tube and Non–G-tube Populations Are Similar

	G-tube Number of Events		No G-tube Number of Events	
TEAE, n (%)	Ganaxolone (n=28)	Placebo (n=19)	Ganaxolone (n=125)	Placebo (n=156)
Infections and infestations	9 (32)	5 (26)	22 (18)	33 (21)
Nervous system disorders	5 (18)	4 (21)	36 (29)	31 (20)
Somnolence/sedation/lethargy	4 (14)	2 (11)	21 (17)	10 (6)
General disorders and administration site conditions	1 (4)	1 (5)	14 (11)	17 (11)
Gastrointestinal disorders	3 (11)	4 (21)	13 (10)	29 (19)
Psychiatric disorders	0	0	9 (7)	12 (8)
Respiratory, thoracic, and mediastinal disorders	5 (18)	0	8 (6)	13 (8)
Metabolism and nutritional disorders	0	0	5 (4)	4 (3)
Eye disorders	0	0	2 (2)	0
Immune system disorders	1 (4)	0	2 (2)	0
Injury and procedural complications	0	0	5 (4)	4 (3)
Investigations	3 (11)	1 (5)	3 (2)	4 (3)
Skin and subcutaneous tissue disorders	1 (4)	3 (16)	2 (2)	5 (3)
Severity				
Mild	17	16	85	118
Moderate	11	3	39	34
Severe	0	0	1	4

All events are reported by system organ class except for somnolence/sedation/lethargy, which are included as preferred terms. Events that were reported more than once in at least 1 group are presented.

Conclusions

- **Based on data from the phase 3 study, efficacy, safety, and drug exposure appear similar between patients administered ganaxolone orally and via g-tube**

References

1. Jakemiec M, et al. *Brain Sci.* 2020;10:107.
2. Olson HE, et al. *Pediatr Neurol.* 2019;97:18-25.

Funding

This study was funded by Marinus Pharmaceuticals, Inc.

Disclosures

All authors are full-time employees of Marinus and hold shares in the company.

Acknowledgment

MedVal Scientific Information Services, LLC provided editorial and graphical support, which was funded by Marinus.

MARINUS
SCIENTIFIC AFFAIRS

Presented at the American Epilepsy Society Annual Meeting, December 1-5, 2023, Orlando, FL